Poster no. PA2-N-036

Scan QR code for poster and supplement

All materials obtained through the QR code are for personal use only and may not be reproduced without permission of the authors

Presenting author:
Alberto M. Pedroncelli

CAM2029 provides long-term and sustained biochemical control of acromegaly: ACROINNOVA 2 Phase 3 final analysis

Joanna L. Spencer-Segal,[1] Julie M. Silverstein,[2] Aleksandra Gilis-Januszewska,[3] Guzin Fidan Yaylali,[4] Elena Isaeva,[5] Mônica R. Gadelha,[6] Beverly M. K. Biller,[7] Pietro Maffei,[8] Maria Fleseriu,[9] Alberto M. Pedroncelli,[10] Jacob Råstam,[10] Maria Harrie,[10] Agneta Svedberg,[10] Fredrik Tiberg,[10] Diego Ferone[11]

[1]Department of Internal Medicine and Michigan Neuroscience Institute, University of Michigan, Ann Arbor, MI, USA; [2]Division of Endocrinology, Metabolism and Lipid Research, Department of Neurosurgery, Washington University School of Medicine, St. Louis, MO, USA; [3]Department of Endocrinology, Jagiellonian University Medical College, Kraków, Poland; [4]Department of Endocrinology and Metabolic Diseases, Faculty of Medicine, Pamukkale University Faculty of Medicine, Denizli, Türkiye; [5]Interregional Clinical Diagnostic Center, Kazan, Russian Federation; [6]Neuroendocrinology Research Center/Endocrinology Division, Medical School and Hospital Universitario Clementino Fraga Filho, Universidade Federal do Rio de Janeiro, Rio de Janeiro, Brazil; [7]Neuroendocrine and Pituitary Tumor Clinical Center, Massachusetts General Hospital, Boston, MA, USA; [8]Department of Medicine, Padua University Hospital, Padua, Italy; [9]Pituitary Center, Departments of Medicine and Neurological Surgery, Oregon Health and Science University, Portland, OR, USA; [10]Camurus AB, Lund, Sweden; [11]Endocrinology Unit, Department of Internal Medicine, IRCCS Ospedale Policlinico San Martino, Genova, Italy

BACKGROUND

- Overproduction of growth hormone (GH) and insulin-like growth factor I (IGF-I) in acromegaly leads to substantial morbidity and significantly reduced quality of life;[1] therefore, effective biochemical control of these hormones is essential to alleviate symptoms and the associated comorbidities of acromegaly[1]
- Injectable first-generation somatostatin receptor ligands (SRLs; octreotide long-acting repeatable [LAR] or lanreotide Autogel [ATG]) can provide effective biochemical control,[2] but patients may experience a substantial treatment burden[3–5]
- CAM2029 is a novel octreotide subcutaneous depot (based on FluidCrystal® technology) with a long-acting formula for convenient monthly self-administration via a ready-to-use autoinjector pen with small-gauge needle[6,7] (see **Supplementary Figure 1**, available via the QR code)
- In a 24-week, Phase 3, randomised, double-blind controlled trial (ACROINNOVA 1; NCT04076462), CAM2029 achieved superior IGF-I control versus placebo (72.2% vs 37.5% of patients, respectively, with IGF-I ≤ upper limit of normal per age and sex [ULN]; *P*=0.0018) and controlled symptoms in patients with IGF-I ≤ULN on standard-of-care (SoC) treatment (octreotide LAR/lanreotide ATG) at screening[6]
- ACROINNOVA 2 (NCT04125836) was a 52-week, Phase 3, open-label trial in which the long-term safety and efficacy of CAM2029 were assessed in patients with acromegaly previously receiving first-generation SRLs (current SoC)
 - Assessments of biochemical and symptom control during the 52-week core phase are reported for the full data set; for corresponding analyses relating to patient-reported outcomes, please see poster PA3-N-004
 - The 52-week extension phase is also complete

CONCLUSIONS

Long-term CAM2029 was well tolerated, with safety findings consistent with those of first-generation injectable SRLs[8]

CAM2029 provided long-term biochemical control for up to 52 weeks among patients who were biochemically controlled on first-generation SRLs. Biochemical control was:
- **Improved in the overall population**
- **Improved among patients directly enrolled into ACROINNOVA 2 (IGF-I ≤2×ULN at screening)**
- **Sustained among patients who had previously achieved control**
- **Regained among patients who received placebo during the ACROINNOVA 1 trial (IGF-I ≤ULN at screening)**

Symptom burden progressively reduced over 52 weeks of CAM2029 treatment

These long-term findings support CAM2029 as an effective and well-tolerated new treatment for acromegaly

METHODS

Analyses for the core phase of ACROINNOVA 2 were undertaken at week 52

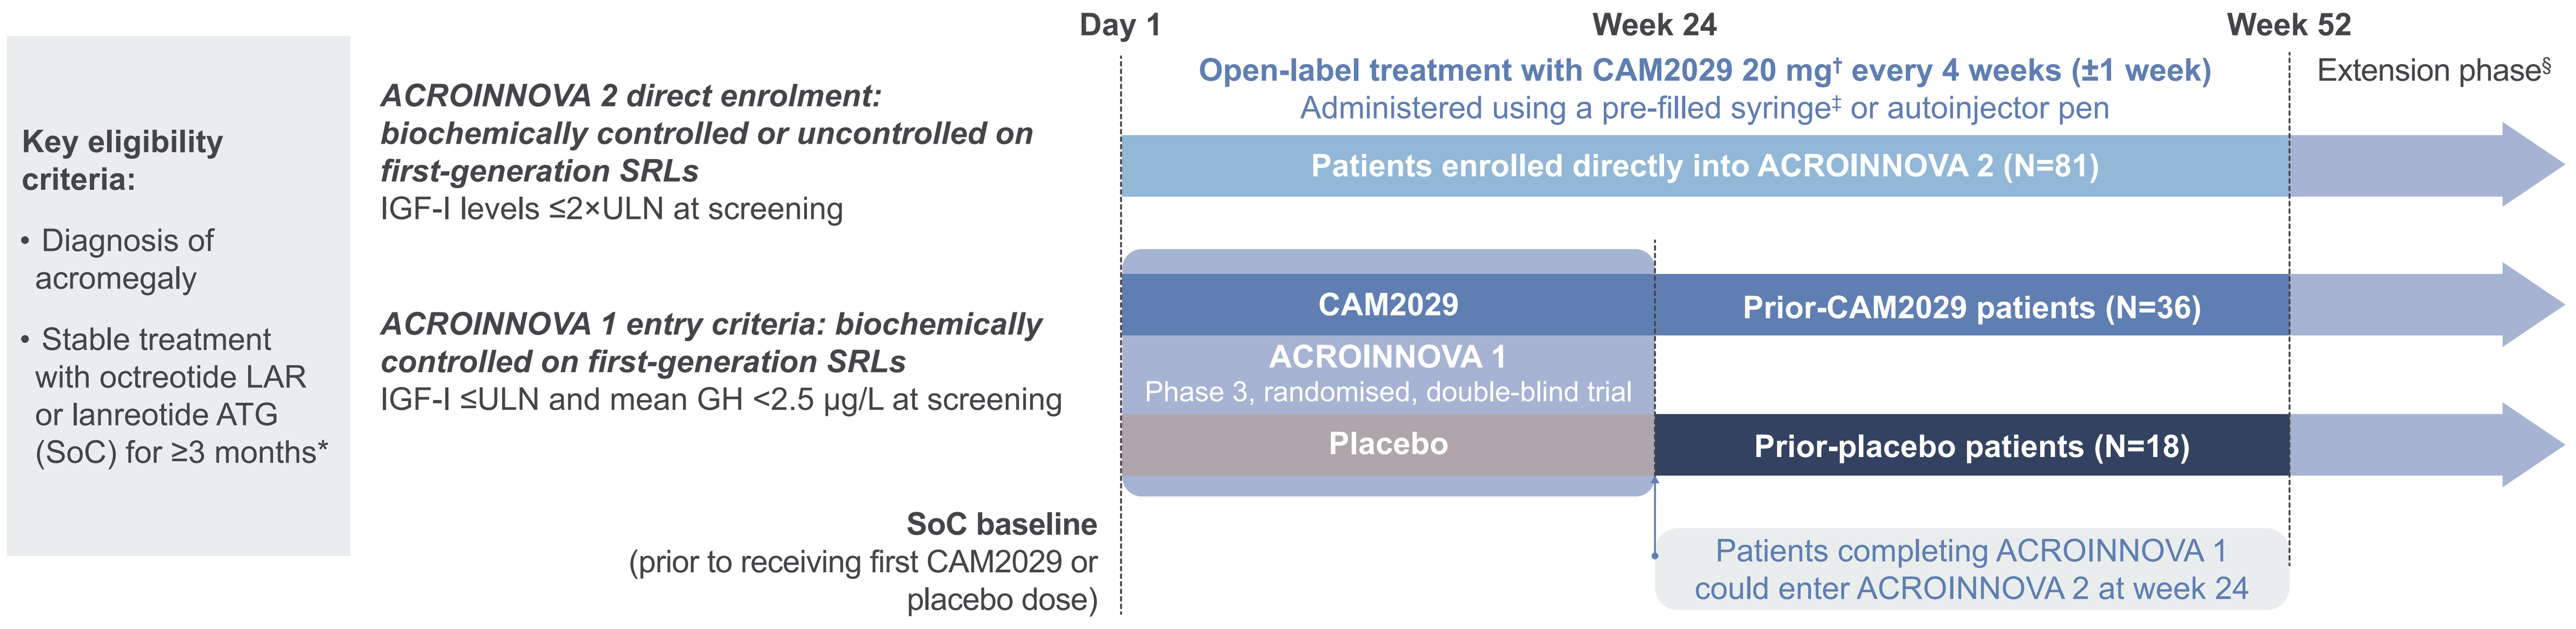

Key eligibility criteria:
• Diagnosis of acromegaly
• Stable treatment with octreotide LAR or lanreotide ATG (SoC) for ≥3 months*
ACROINNOVA 2 direct enrolment: biochemically controlled or uncontrolled on first-generation SRLs
IGF-I levels ≤2×ULN at screening
ACROINNOVA 1 entry criteria: biochemically controlled on first-generation SRLs
IGF-I ≤ULN and mean GH <2.5 μg/L at screening
SoC baseline
(prior to receiving first CAM2029 or placebo dose)
Day 1
Week 24
Week 52
Open-label treatment with CAM2029 20 mg† every 4 weeks (±1 week)
Administered using a pre-filled syringe‡ or autoinjector pen
Extension phase§
Patients enrolled directly into ACROINNOVA 2 (N=81)
CAM2029
ACROINNOVA 1
Phase 3, randomised, double-blind trial
Placebo
Prior-CAM2029 patients (N=36)
Prior-placebo patients (N=18)
Patients completing ACROINNOVA 1 could enter ACROINNOVA 2 at week 24

*Treatment with a stable dose of octreotide LAR (10, 20, 30 or 40 mg) or lanreotide ATG (60, 90 or 120 mg); †If required based on safety and tolerability, the dose could be reduced to 10 mg CAM2029; ‡Only the autoinjector pen is available following marketing authorisation;[9] §Upon completing the core phase in ACROINNOVA 2, patients could continue to receive CAM2029 in a 52-week extension phase.

Primary endpoint
• Characterisation of adverse events (AEs)
Selected secondary endpoints
Biochemical control
• Proportion of patients with mean IGF-I ≤ULN (average of week 50 and 52 measurements)
Symptoms of acromegaly*
• Change in Acromegaly Index of Severity (AIS)* score from baseline to week 52

*Details for AIS are provided in **Supplementary Figure 2**, available via the QR code.

RESULTS

Overall, 127 patients (94.1%) completed the core phase in ACROINNOVA 2

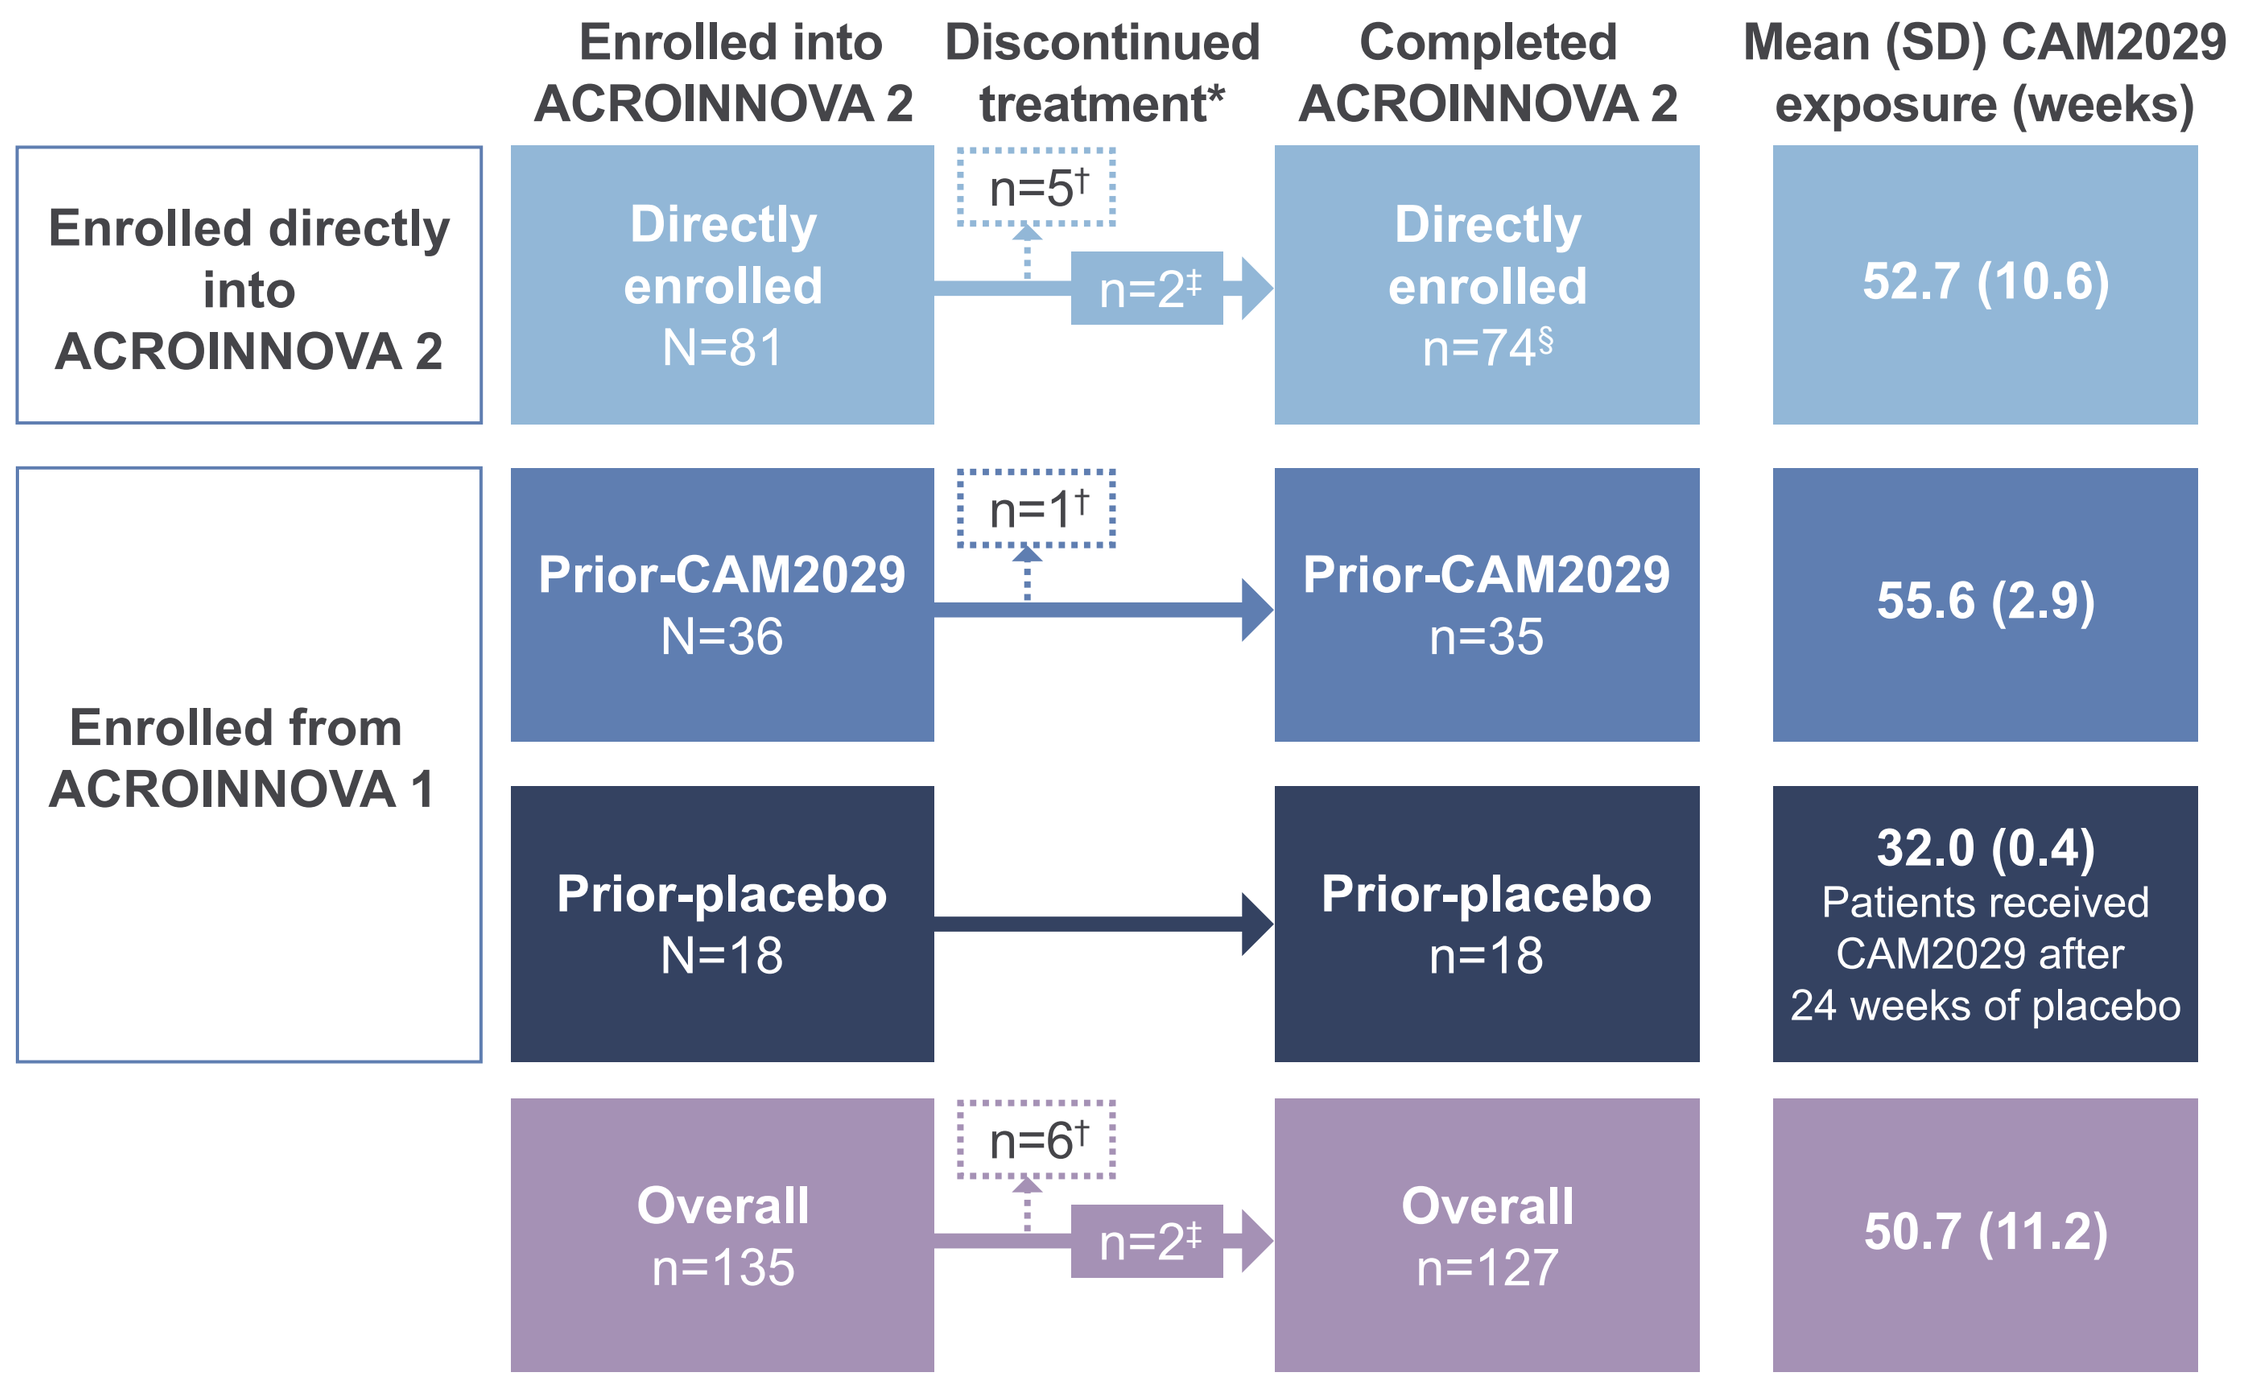

Enrolled into ACROINNOVA 2
Discontinued treatment*
Completed ACROINNOVA 2
Mean (SD) CAM2029 exposure (weeks)
Enrolled directly into ACROINNOVA 2
Directly enrolled N=81
n=5†
n=2‡
Directly enrolled n=74§
52.7 (10.6)
Enrolled from ACROINNOVA 1
Prior-CAM2029 N=36
n=1†
Prior-CAM2029 n=35
55.6 (2.9)
Prior-placebo N=18
Prior-placebo n=18
32.0 (0.4)
Patients received CAM2029 after 24 weeks of placebo
Overall n=135
n=6†
n=2‡
Overall n=127
50.7 (11.2)

Data shown are for patients completing the 52-week core phase of ACROINNOVA 2. *Reasons for discontinuation: consent withdrawn (n=5, directly enrolled group); AEs (directly enrolled group, n=2: injection site haemorrhage [Grade 1] and depression [Grade 1]); patient declined to attend study site (prior-CAM2029 group, n=1); †Discontinued and withdrew from trial; ‡Patients discontinued treatment but were followed up to the end of the trial; §Two patients completed treatment with CAM2029 but did not attend the week 52 visit. SD, standard deviation.

- Patient demographics and medical histories were collected at screening, see **Supplementary Table 1** (available via QR code)

CAM2029 was well tolerated, most AEs were mild and no new safety signals were observed up to week 52

n (%)	Directly enrolled (N=81)	Prior-CAM2029 (N=36)	Prior-placebo (N=18)	Overall (N=135)
Any AE	56 (69.1)	34 (94.4)	12 (66.7)	102 (75.6)
Any CAM2029-related AE	39 (48.1)	23 (63.9)	12 (66.7)	74 (54.8)
Any AE of Grade				
1	51 (63.0)	32 (88.9)	11 (61.1)	94 (69.6)
2	22 (27.2)	19 (52.8)	6 (33.3)	47 (34.8)
3	9 (11.1)	7 (19.4)	1 (5.6)	17 (12.6)
Any SAE	6 (7.4)	8 (22.2)	1 (5.6)	15 (11.1)
Any CAM2029-related SAE	0	0	1 (5.6)*	1 (0.7)
Any AE leading to discontinuation of CAM2029	2 (2.5)	0	0	2 (1.5)
Any AE leading to dose reduction	0	0	1 (5.6)	1 (0.7)
Any injection site AE†	32 (39.5)	19 (52.8)	9 (50.0)	60 (44.4)

AEs refer to TEAEs. Only TEAEs and SAEs reported from the first administration of CAM2029 until week 52 are included. *Cholelithiasis (Grade 2; resolved); †AEs included injection site bruising, dermatitis, discomfort, erythema, extravasation, haematoma, haemorrhage, hypertrophy, inflammation, induration, mass, nodule, oedema, pain, pruritus, reaction and swelling. SAE, serious AE; TEAE, treatment-emergent AE.

- All injection site AEs were mild (Grade 1) or moderate (Grade 2)
 - New injection site AEs generally decreased with increasing injection number during CAM2029 treatment in all groups (**Supplementary Figure 3**, available via QR code)
- The most common AEs (≥10% of patients) in the overall population were injection site erythema (27.4%), COVID-19 and injection site swelling (each 14.8%), headache (14.1%) and arthralgia (12.6%)
 - For AEs by preferred term and patient group, reported in ≥5% of patients in any group and ≥3 patients overall, see **Supplementary Table 2**, available via QR code

CAM2029 provided effective long-term biochemical control

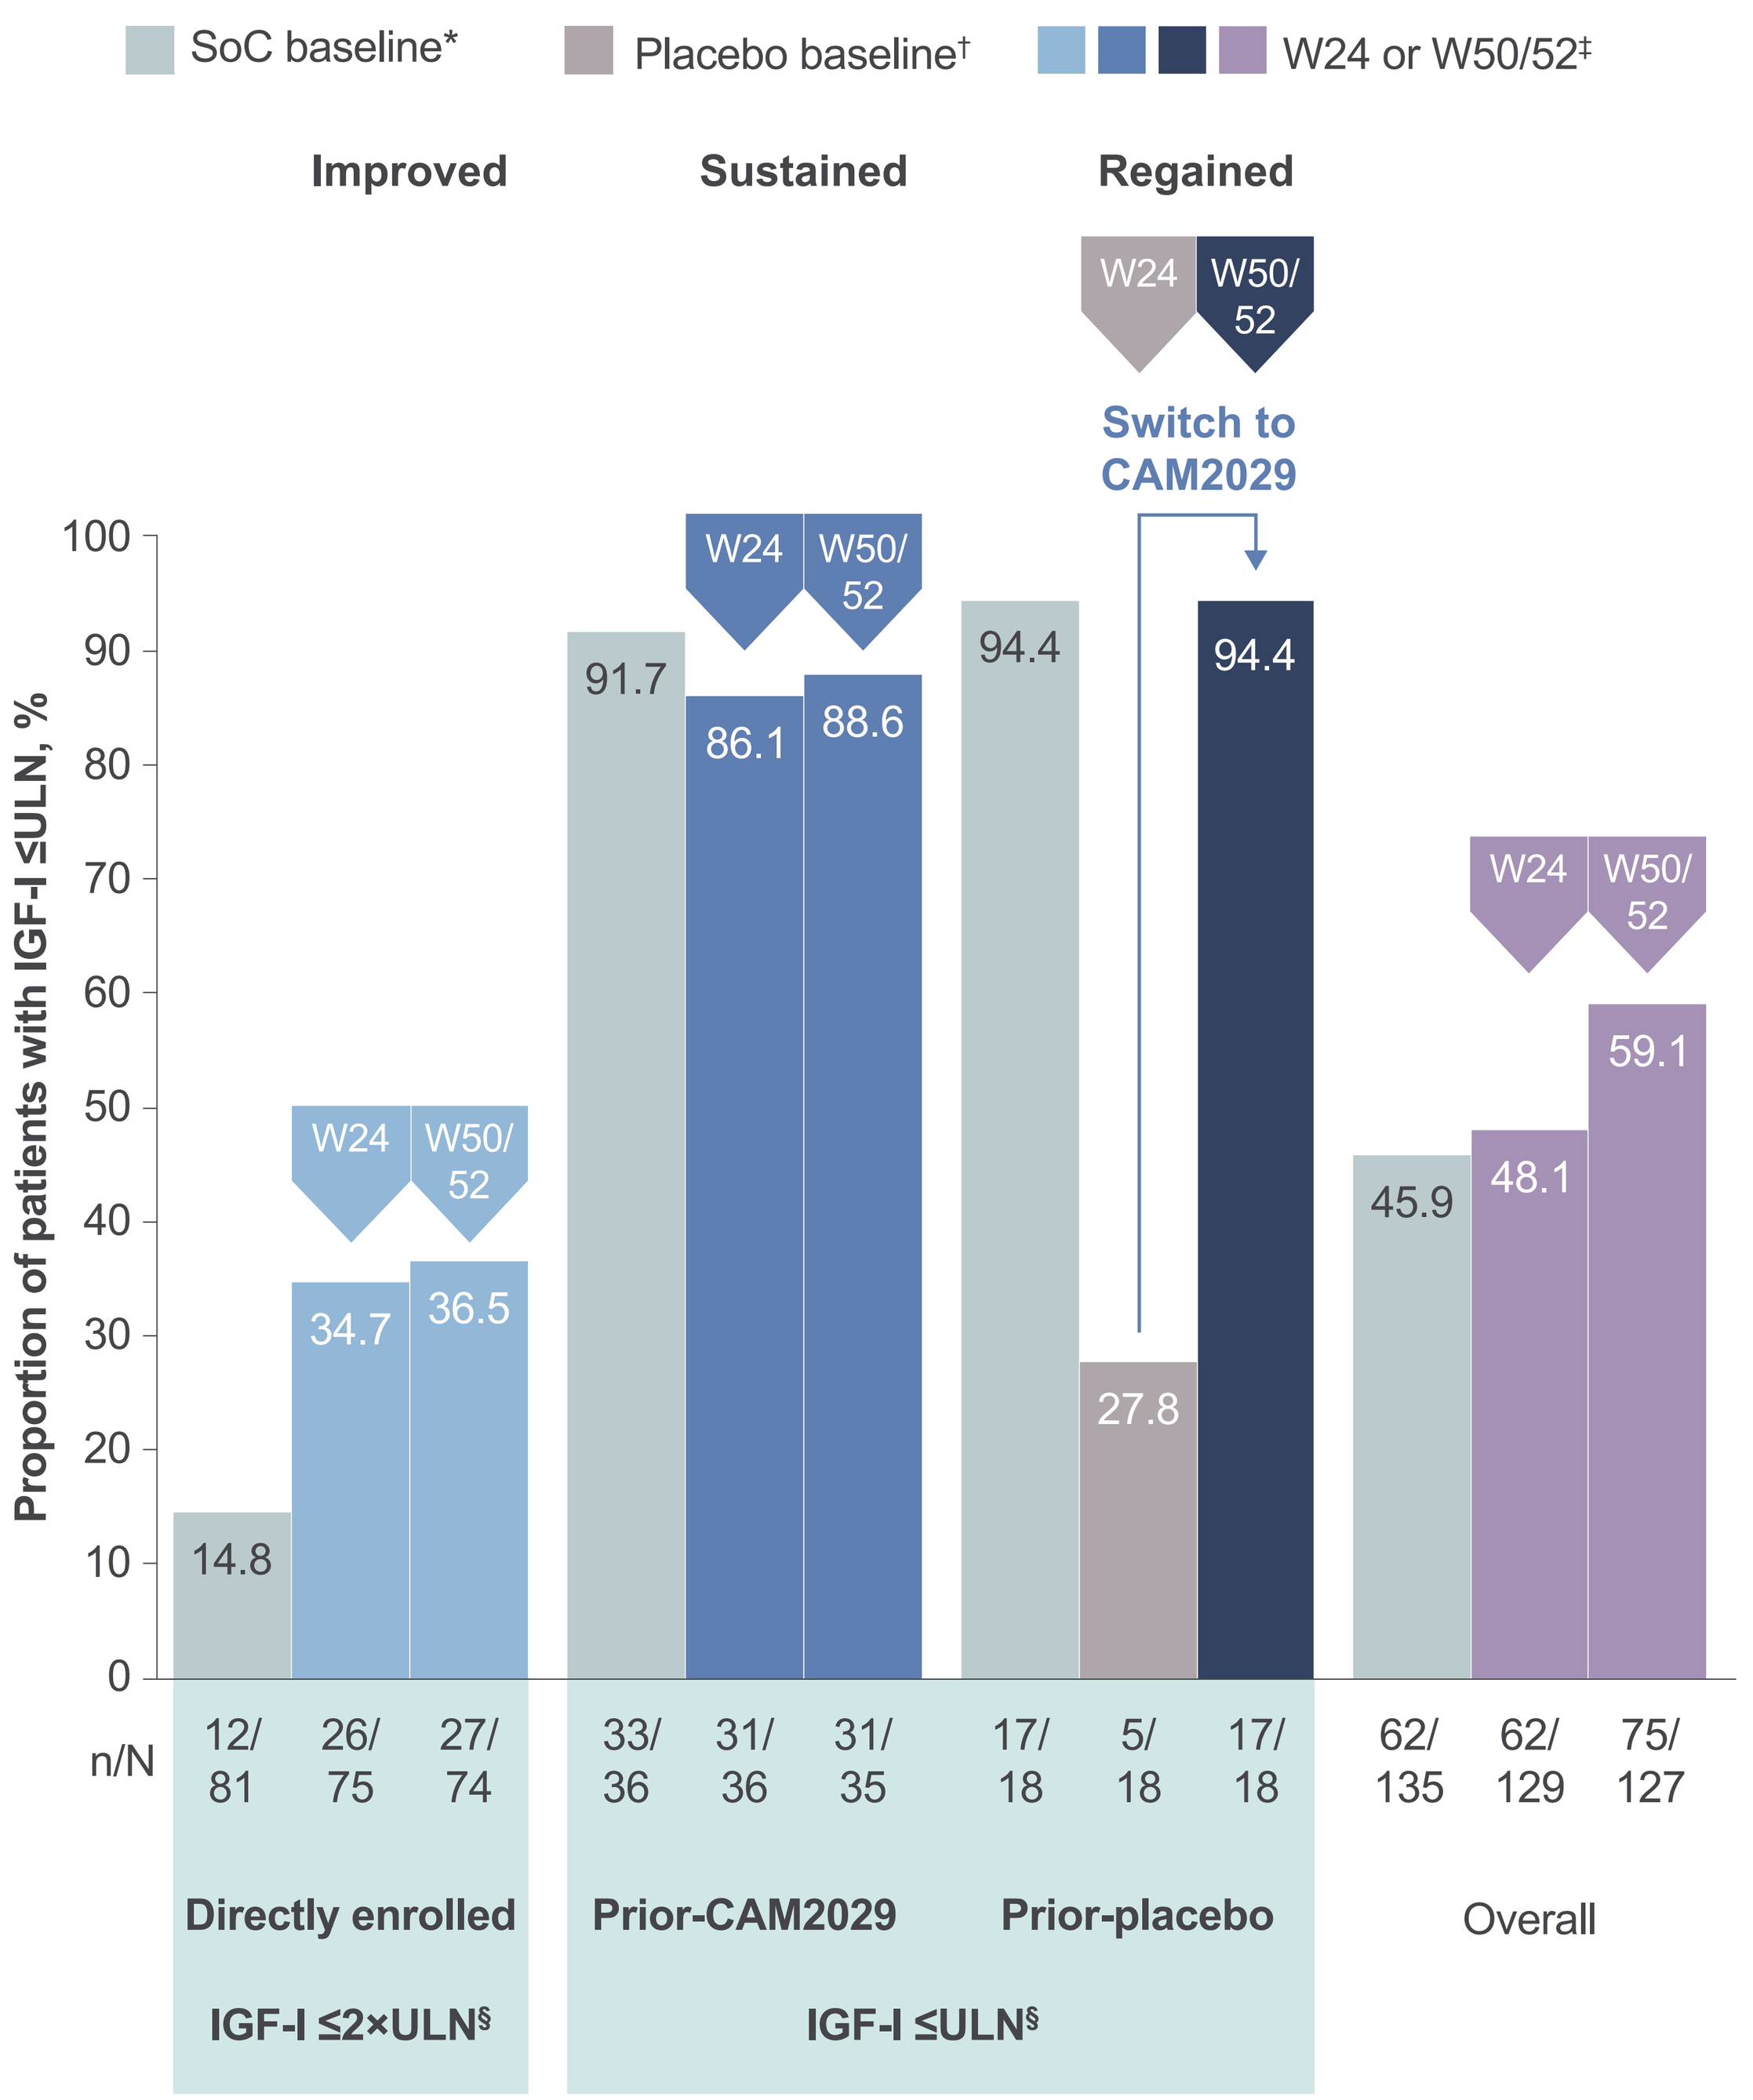

SoC baseline*
Placebo baseline†
W24 or W50/52‡
Improved
Sustained
Regained
Switch to CAM2029
Proportion of patients with IGF-I ≤ULN, %
100
90
80
70
60
50
40
30
20
10
0
W24
W50/52
14.8
34.7
36.5
91.7
86.1
88.6
94.4
27.8
94.4
45.9
48.1
59.1
n/N
12/81
26/75
27/74
33/36
31/36
31/35
17/18
5/18
17/18
62/135
62/129
75/127
Directly enrolled
Prior-CAM2029
Prior-placebo
Overall
IGF-I ≤2×ULN§
IGF-I ≤ULN§

Figure is based on all patients with available data at the timepoint. *Before receiving the first CAM2029/placebo dose; IGF-I values are means from assessments at week −2 and day 1; †Prior-placebo group includes one patient switched to SoC from placebo during ACROINNOVA 1; IGF-I value is the mean from assessments at weeks 22 and 24; ‡IGF-I values are means from assessments at weeks 50 and 52; §At screening. W, week.

Mean IGF-I levels were stable in the overall population during CAM2029 treatment

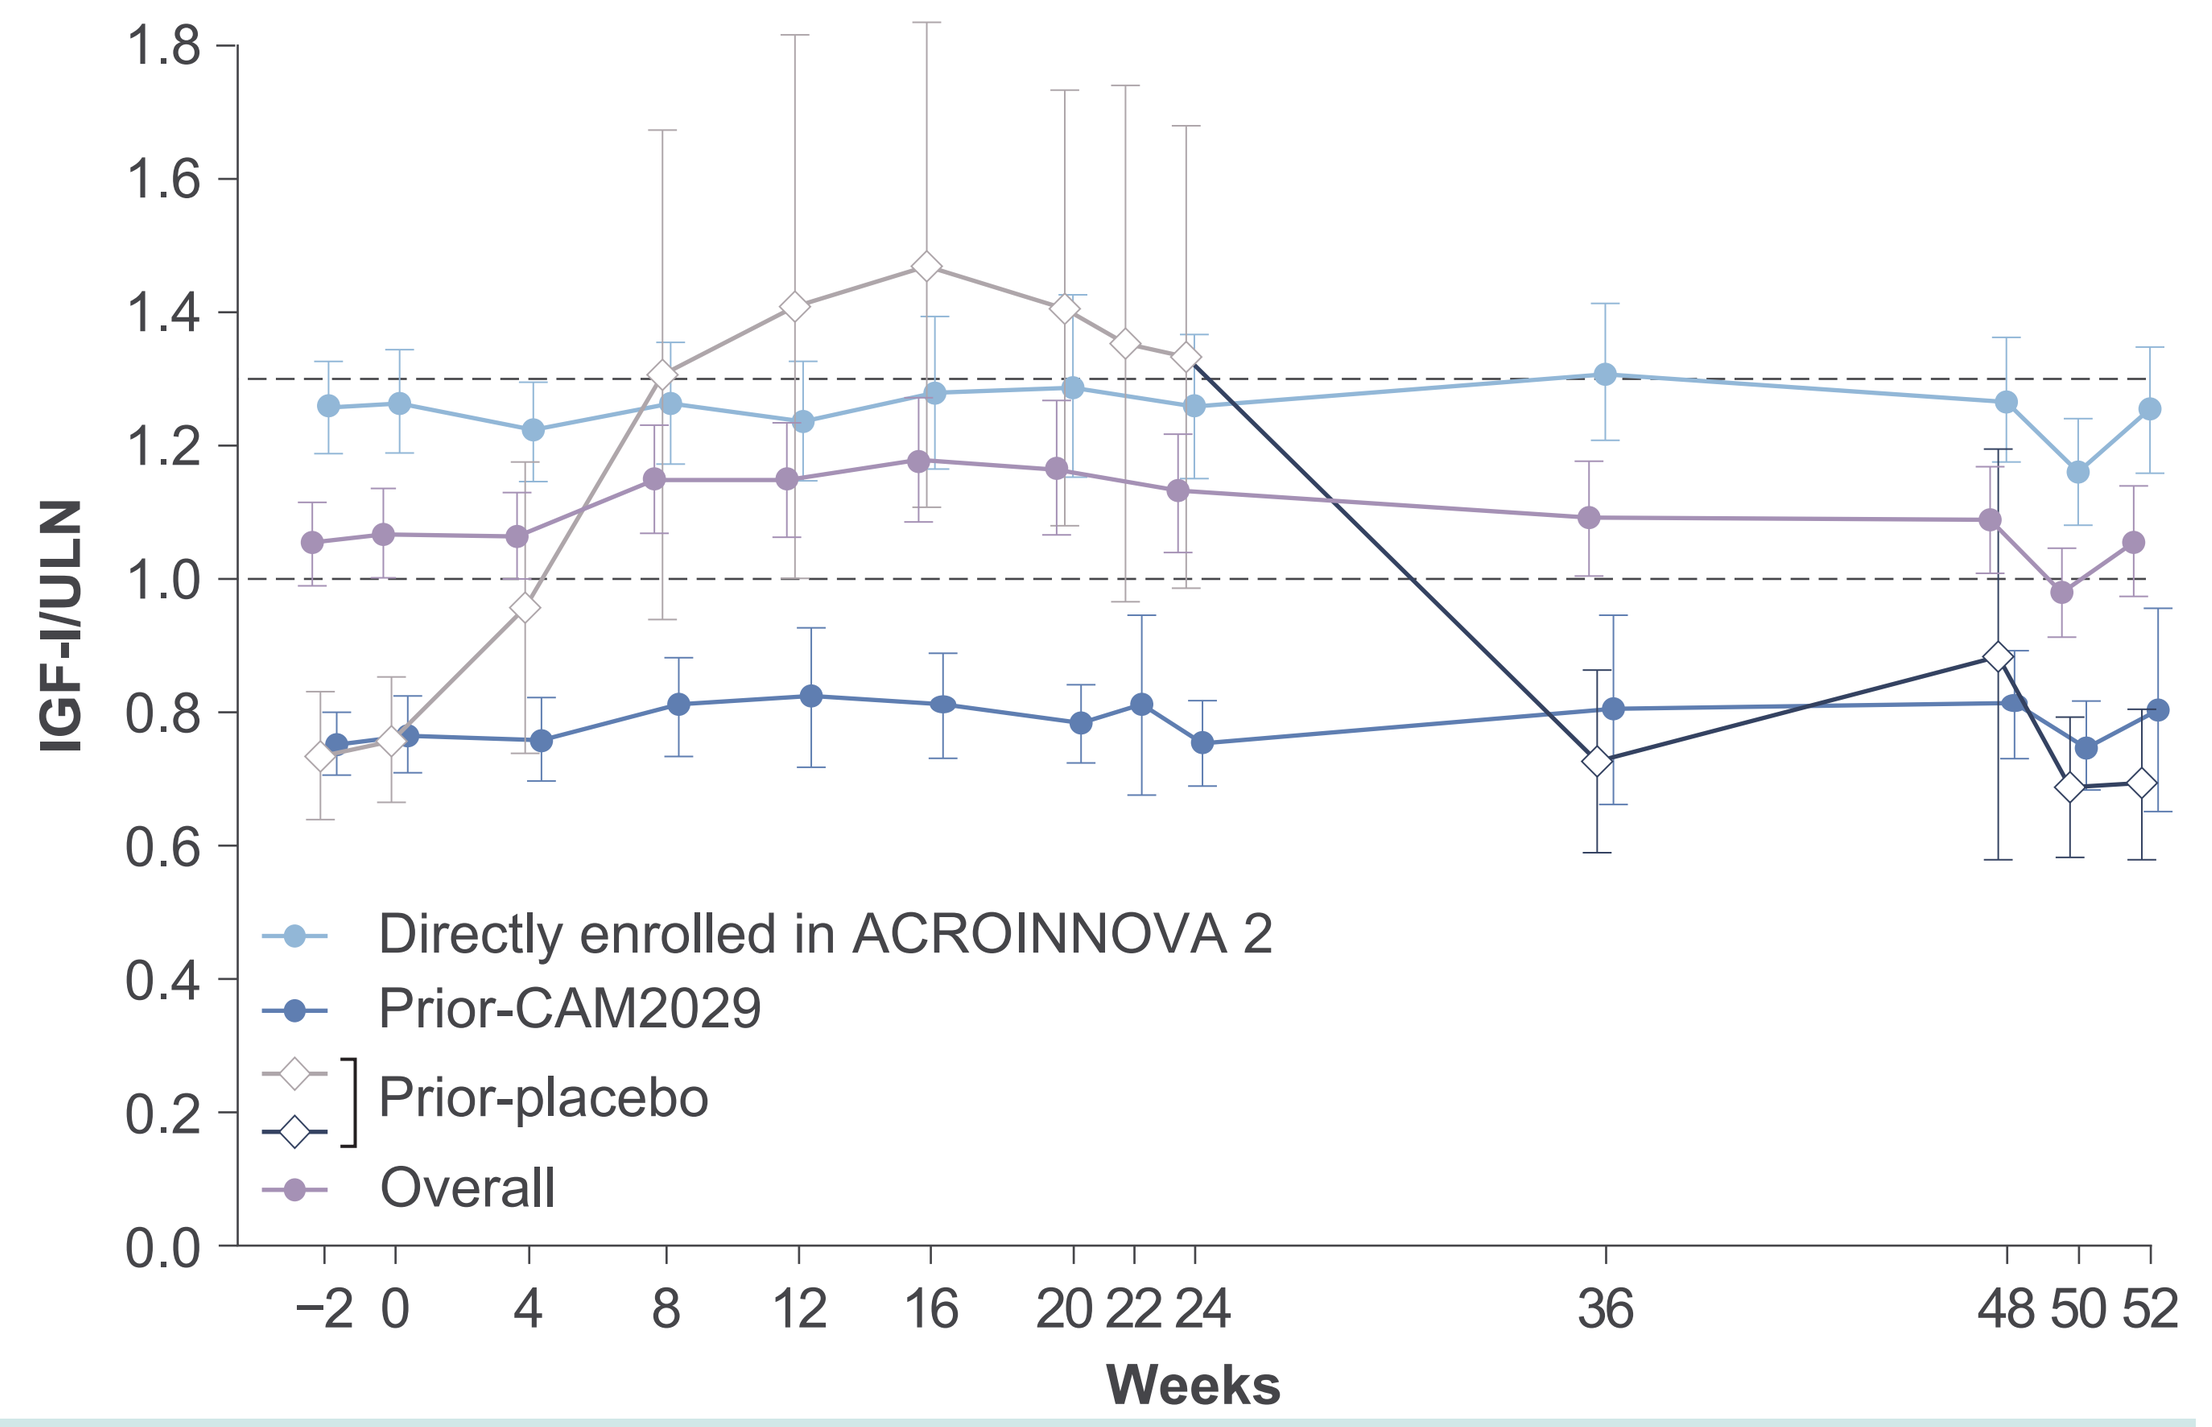

IGF-I/ULN
1.8
1.6
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0.0
Directly enrolled in ACROINNOVA 2
Prior-CAM2029
Prior-placebo
Overall
−2
0
4
8
12
16
20
22
24
36
48
50
52
Weeks

n	−2	0	4	8	12	16	20	22	24	36	48	50	52
Directly enrolled	81	81	80	80	80	79	78	NA	75	74	74	69	74
Prior-CAM2029	36	35	35	35	36	36	36	35	36	35	35	33	33
Prior-placebo	18	18	18	18	18	18	18	17	18	17	18	17	17
Overall	135	134	133	133	134	133	132	NA	129	126	127	119	124

Data are from the intention-to-treat population. Data shown include the 24-week placebo period before the prior-placebo group was switched to CAM2029 (pale grey diamonds); these patients are also included in the overall population. Error bars represent 95% confidence interval (CI). Dashed horizontal rules represent ULN and 1.3×ULN.

Symptoms continuously improved from baseline to week 52 in the overall population

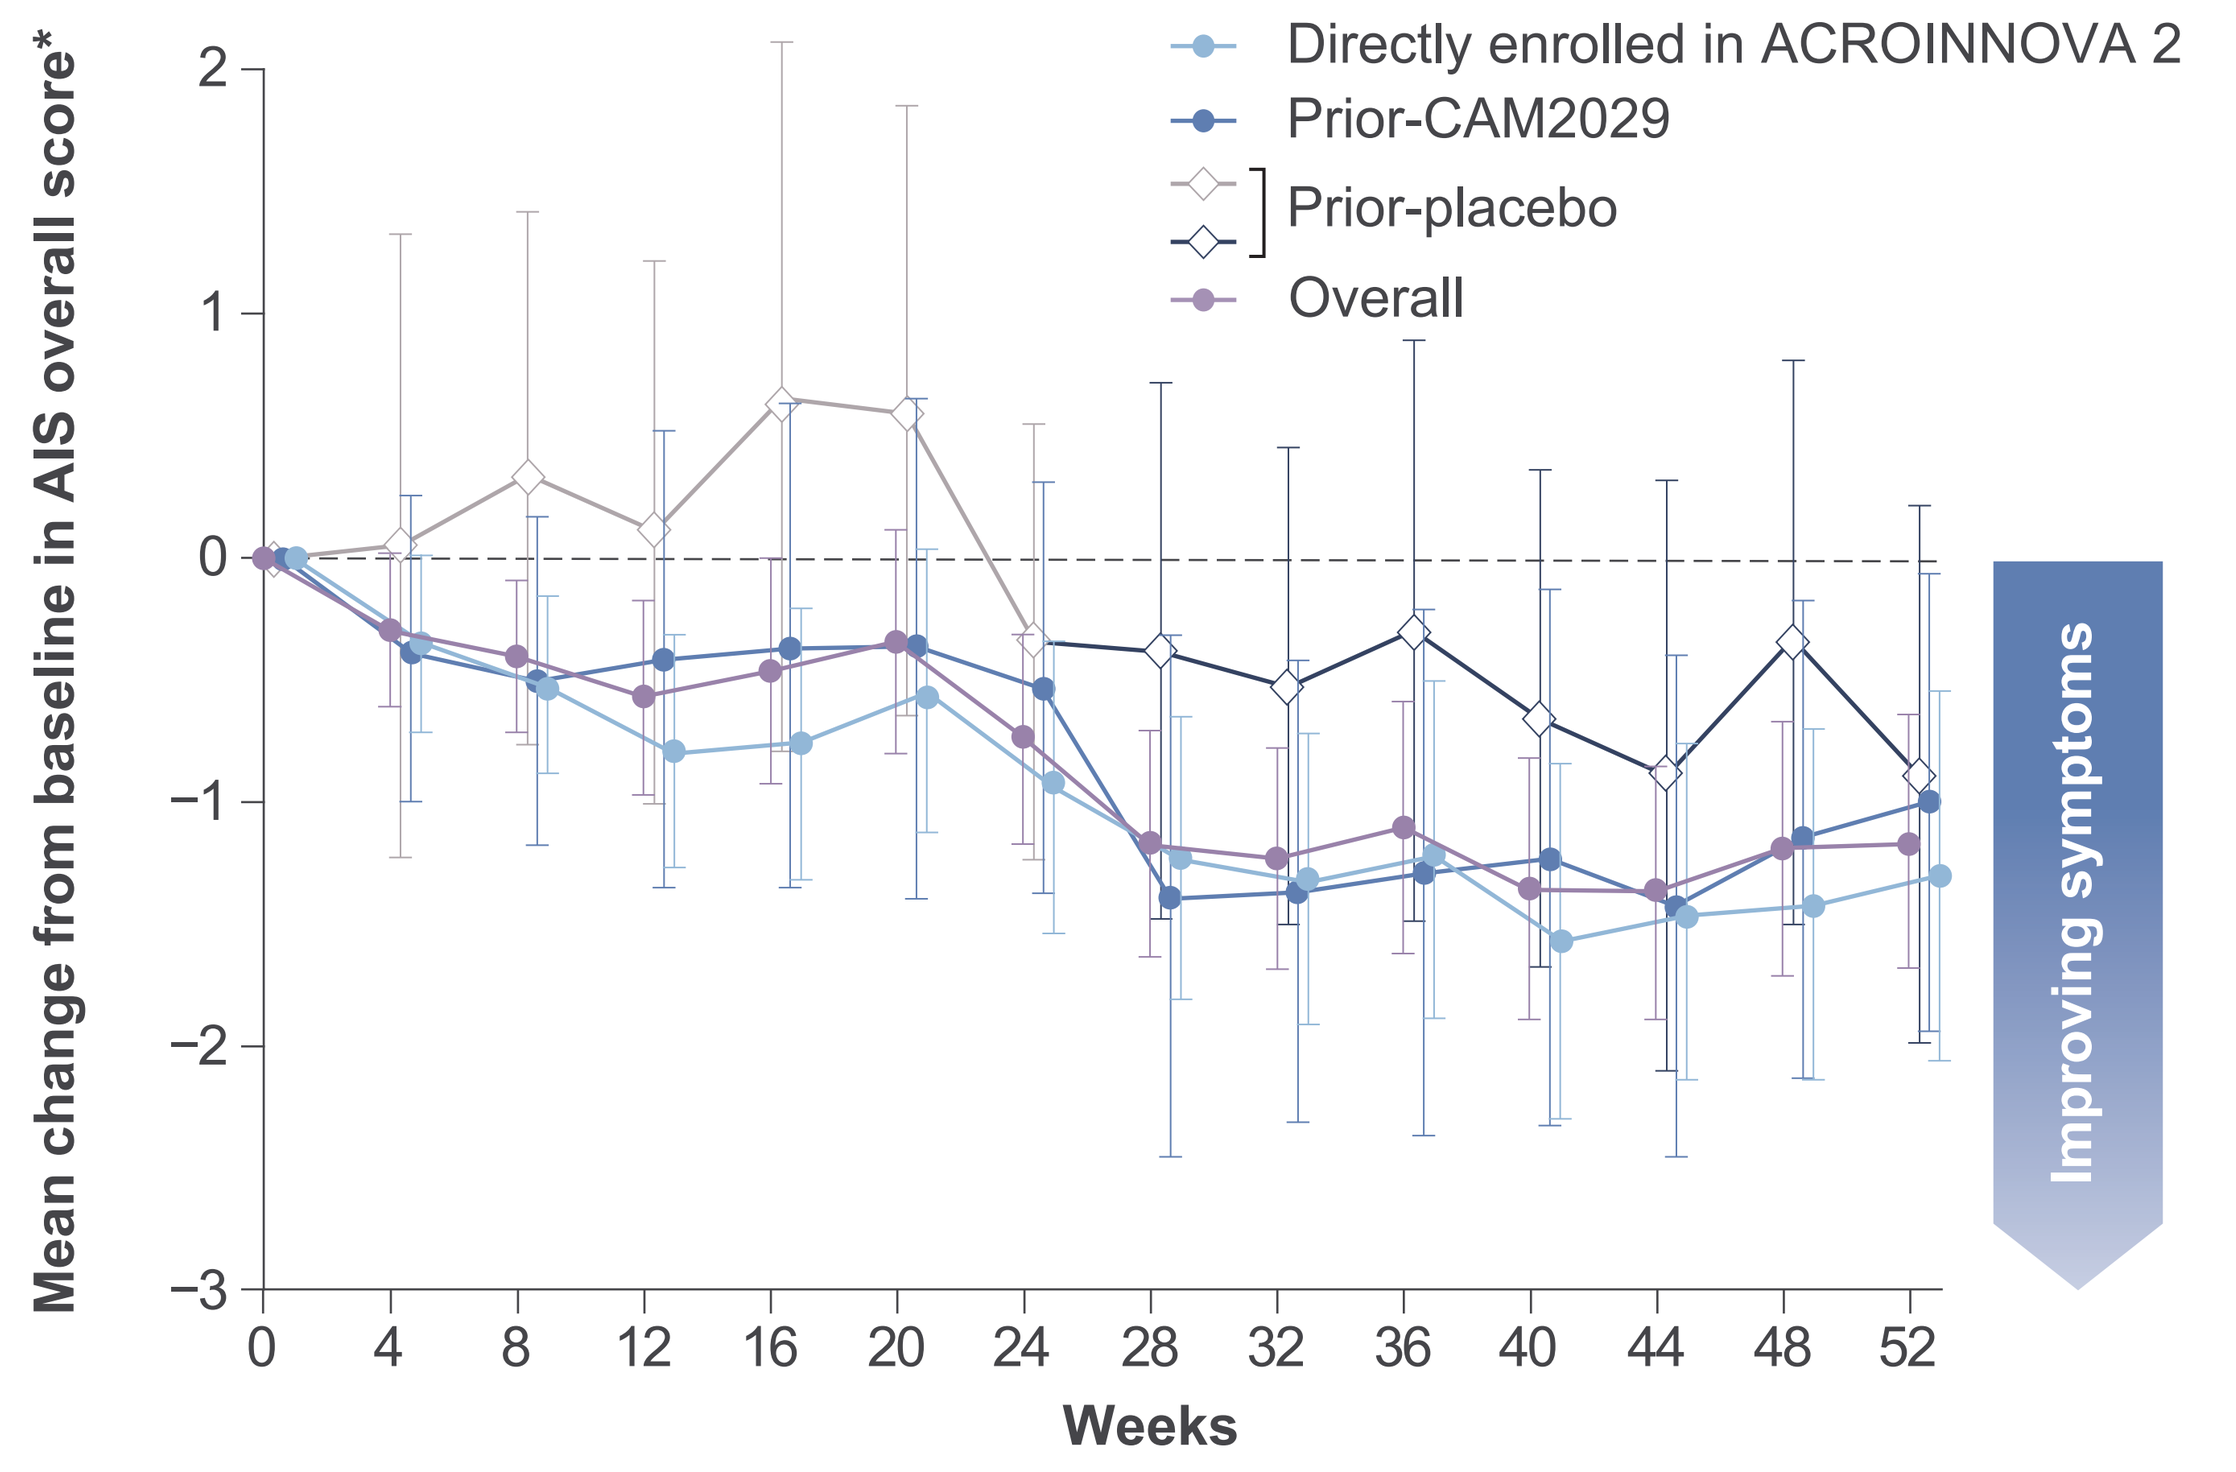

Mean change from baseline in AIS overall score*
2
1
0
−1
−2
−3
Directly enrolled in ACROINNOVA 2
Prior-CAM2029
Prior-placebo
Overall
Improving symptoms
0
4
8
12
16
20
24
28
32
36
40
44
48
52
Weeks

Data include the 24-week placebo period before the prior-placebo group switched to CAM2029 (pale grey diamonds); the group is also included in the overall population. *AIS 0–18; sum of scores (0–3; none–severe) for six symptoms (headache, sweating, fatigue, joint pain, paraesthesia and soft tissue swelling). Error bars represent 95% CI.

- In the overall population at week 52, it was estimated that 10.9% fewer patients had any acromegaly symptoms than at SoC baseline (95% CI: –17.1, –4.7; n=124)

References

1. Giustina A *et al. J Clin Endocrinol Metab* 2020;105:e937–46; **2.** Melmed S *et al. Nat Rev Endocrinol* 2018;14:552–61; **3.** Geer EB *et al. BMC Endocrine Disord* 2020;20:117; **4.** Fleseriu M *et al. Front Endocrinol (Lausanne)* 2021;12:627711; **5.** Gurel MH *et al. Patient Prefer Adherence* 2014;8:53–62; **6.** Ferone D *et al. J Clin Endocrinol Metab* 2025;110:1729–39; **7.** Tiberg F *et al. Br J Clin Pharmacol* 2015;80:460–72; **8.** Fleseriu M *et al. Lancet Diabetes Endocrinol* 2022;10:102–11; **9.** Camurus AB. Oczyesa® (CAM2029). EU summary of product characteristics, 2025.

Acknowledgements and disclosures

The ACROINNOVA 2 trial was funded by Camurus AB. Thank you to the patients, investigators, nurses and trial coordinators who made this trial possible. Medical writing assistance was provided by Lisa Heaney, PhD, at Amiculum® and was funded by Camurus AB.

COI disclosures: AMP is employed by Camurus AB (CAB), was previously employed by Recordati Rare Diseases (RRD), and holds stock in CAB, Novartis and RRD. JLSS and MRG have served as advisors to CAB and Crinetics Pharmaceuticals (CT), with JLSS also advising Corcept Therapeutics and MRG also advising RRD and Novo Nordisk (NN). JLSS received fees from CAB and research funding from CAB, RRD and Alexion Pharmaceuticals (AP). JMS received fees from CAB and Xeris and research funding from DebioPharm, Xeris, Lundbeck, NN and AbbVie. AGJ received fees from Recordati. MRG received fees and research funding from CT and RRD and fees from CAB and NN. BMKB received consulting honoraria from Ascendis, CAB, Chiesi, CT, Lundbeck, Neurocrine, NN, Pfizer, RRD and Xeris and institutional research grants from Ascendis, NN and CT. PM received research funding from RRD. MF received fees from AP, CAB, CT, DebioPharm and RRD and research funding from AP, CT and DebioPharm. JR, MH AS and FT are employed by CAB and hold stock in CAB. AS has served as an advisor to Pharma Holdings AS. GFY and EI have nothing to declare.

Presented at the 22nd International Congress of Endocrinology (ICE); Kyoto, Japan; 2–5 June 2026. Previously presented at the Joint Congress of the European Society for Paediatric Endocrinology and the European Society of Endocrinology (ESPE/ESE); Copenhagen, Denmark; 10–13 May 2025, the Endocrine Society's Annual Meeting (ENDO); San Francisco, CA, USA; 12–15 July 2025, the Deutsche Gesellschaft für Endokrinologie (DGE) Herbstsitzung der AG Hypophyse und 28. Jahrestagung der Sektion Neuroendokrinologie (Joint Meeting); Hamburg, Germany; 9–10 October 2025 and the and the Workshop of the European Neuroendocrine Association (ENEA) Marseille, France; 3–5 December 2025.