Poster no.
PA1-N-028

Scan QR code
for poster and supplement

All materials obtained through the QR code are for personal use only and may not be reproduced without permisson of the authors

Presenting author: Alberto M. Pedroncelli

CAM2029 maintains IGF-I and acromegaly symptom control across the 4-week dosing interval and intervals >28 days: data from ACROINNOVA 1

Mônica R. Gadelha,[1] Beverly M. K. Biller,[2] Maria Fleseriu,[3] Joanna L. Spencer-Segal,[4] Julie M. Silverstein,[5] Irina Bancos,[6] Jacob Råstam,[7] Alberto M. Pedroncelli,[7] Fredrik Tiberg,[7] Diego Ferone[8]

[1]Neuroendocrinology Research Center/Endocrinology Division, Medical School and Hospital Universitario Clementino Fraga Filho, Universidade Federal do Rio de Janeiro, Rio de Janeiro, Brazil; [2]Neuroendocrine and Pituitary Tumor Clinical Center, Massachusetts General Hospital, Boston, MA, USA; [3]Pituitary Center, Departments of Medicine and Neurological Surgery, Oregon Health and Science University, Portland, OR, USA; [4]Department of Internal Medicine and Michigan Neuroscience Institute, University of Michigan, Ann Arbor, MI, USA; [5]Division of Endocrinology, Metabolism and Lipid Research, Department of Neurosurgery, Washington University School of Medicine, St. Louis, MO, USA; [6]Division of Endocrinology, Mayo Clinic, Rochester, MN, USA; [7]Camurus AB, Lund, Sweden; [8]Endocrinology Unit, Department of Internal Medicine, IRCCS Ospedale Policlinico San Martino, Genova, Italy

BACKGROUND

- Acromegaly is characterised by the overproduction of growth hormone (GH) and insulin-like growth factor I (IGF-I), leading to substantial morbidity and reduced quality of life (QoL)[1]
- Biochemical control (IGF-I ≤upper limit of normal [ULN] per age)[2,3] can be provided by standard-of-care (SoC) medical treatment (first-generation injectable somatostatin receptor ligands [SRLs]; octreotide long-acting repeatable [LAR] or lanreotide Autogel [ATG])[4]
- For patients with acromegaly, symptom control is a priority.[5] However, some patients report worsening or re-emergence of symptoms towards the end of the SoC monthly dosing interval[6,7]
- CAM2029 is a novel octreotide subcutaneous depot (based on FluidCrystal® technology) with a long-acting formula for convenient monthly self-administration via a ready-to-use pre-filled pen (autoinjector) with a small-gauge needle[8,9] (see **Supplementary Figure 1**, available via the QR code)
- ACROINNOVA 1 was a 24-week, Phase 3, randomised, double-blind, placebo-controlled trial (NCT04076462) of once-monthly CAM2029 (±1 week) in patients with IGF-I ≤ULN per age and sex while receiving SoC treatment at screening[9]
 - CAM2029 achieved superior IGF-I control versus placebo (72.2 vs 37.5% of patients, respectively; *P*=0.0018)
 - Patients receiving CAM2029 had well-controlled symptoms and improved QoL compared with baseline SoC
 - The safety profile was consistent with those of first-generation SRLs

CONCLUSIONS

In patients biochemically controlled at screening on SoC and receiving CAM2029 for 24 weeks, IGF-I control was maintained and symptoms were consistently well managed throughout a 4-week post-dose interval

More than half of patients had later assessments (intervals >28 days); these patients also maintained biochemical and symptom control

These findings provide useful information on the efficacy profile of CAM2029 lasting beyond a 28-day dosing window, reinforcing the potential of CAM2029 to control the burden of disease and address unmet needs among patients with acromegaly

METHODS

The stability of IGF-I and symptom control was evaluated following the final CAM2029 dose in ACROINNOVA 1

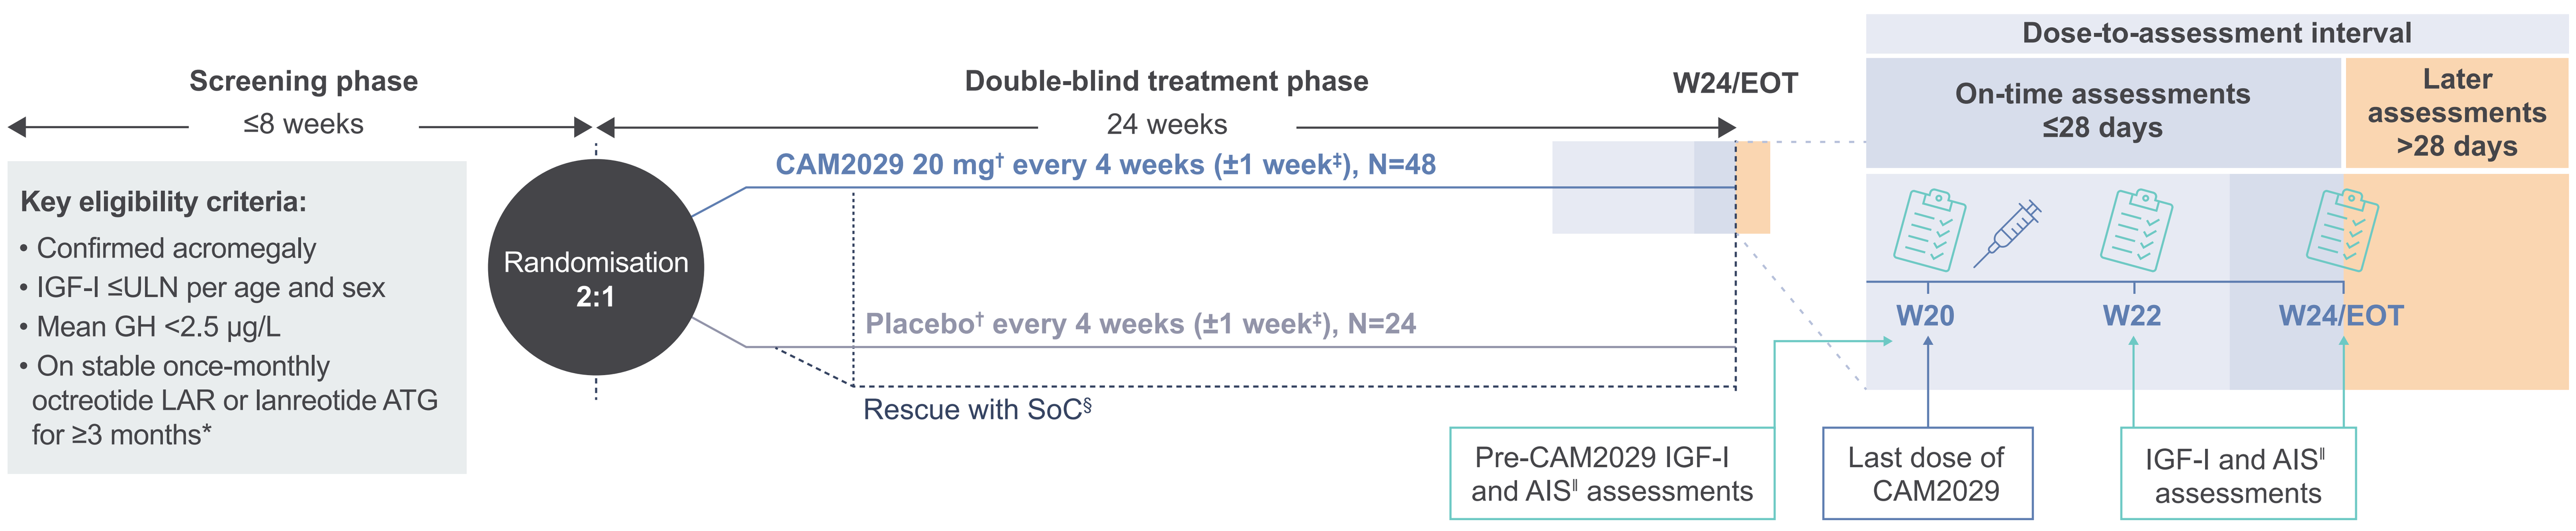
Screening phase
≤8 weeks
Key eligibility criteria:
• Confirmed acromegaly
• IGF-I ≤ULN per age and sex
• Mean GH <2.5 µg/L
• On stable once-monthly octreotide LAR or lanreotide ATG for ≥3 months*
Randomisation
2:1
Double-blind treatment phase
24 weeks
W24/EOT
CAM2029 20 mg† every 4 weeks (±1 week‡), N=48
Placebo† every 4 weeks (±1 week‡), N=24
Rescue with SoC§
Dose-to-assessment interval
On-time assessments
≤28 days
Later assessments
>28 days
W20
W22
W24/EOT
Pre-CAM2029 IGF-I and AIS‖ assessments
Last dose of CAM2029
IGF-I and AIS‖ assessments

- We report descriptive data from W20, W22 and W24/EOT from patients completing their W20 CAM2029 dose and their W24/EOT assessments, with either assessments performed at ≤28 days post-dose (on-time assessments) or assessments performed at >28 days post-dose (later assessments):
 - Proportion of patients with IGF-I ≤ULN (W24/EOT only)
 - IGF-I/ULN values (subgroup means and individual patients)
 - AIS overall scores (subgroup means and individual patients)
 - Proportion of patients with individual symptoms of acromegaly (subgroups)

Adapted from Ferone D *et al. J Clin Endocrinol Metab* 2025;110:1729–39. *Octreotide LAR 10, 20, 30 or 40 mg or lanreotide ATG 60, 90 or 120 mg; †If required, dose reduction to 10 mg CAM2029 or 0.5 mL placebo for safety and tolerability; ‡For patient convenience, administration of CAM2029/placebo was permitted within a window of ±1 week around each scheduled 4-weekly dose. Dose timings were not adjusted in relation to any potential lack of efficacy or safety issue; §Patients who were rescued by switch to SoC continued to participate in the trial and attended all planned visits (n=1, placebo group); ‖Details of the AIS (evaluation of key acromegaly symptoms by clinician and patient together) are provided in **Supplementary Figure 2**, available via the QR code. AIS, Acromegaly Index of Severity; EOT, end of trial; W, week.

RESULTS

- 48 patients were randomised to CAM2029 in ACROINNOVA 1 (see **Supplementary Table 1** for more details, available via the QR code)[9]
- 42 (87.5%) patients completed CAM2029 treatment and formed the population for the analyses reported here[9]
 - Two patients withdrew from the trial and four discontinued treatment because of adverse events
- W24/EOT assessments were performed ≤28 days post-dose (on time) for 19 (45.2%) patients and were later (>28 days post-dose) for 23 (54.8%) patients (including six patients with assessments delayed >35 days post-dose). Some later assessments were due to COVID-19
- The mean (range) duration of the final dosing interval was:
 - 25.7 (21–28) days for the on-time assessment group
 - 33.4 (29–42) days for the later assessment group

Most patients achieved biochemical control at W24/EOT, including those with assessments performed >28 days post-dose

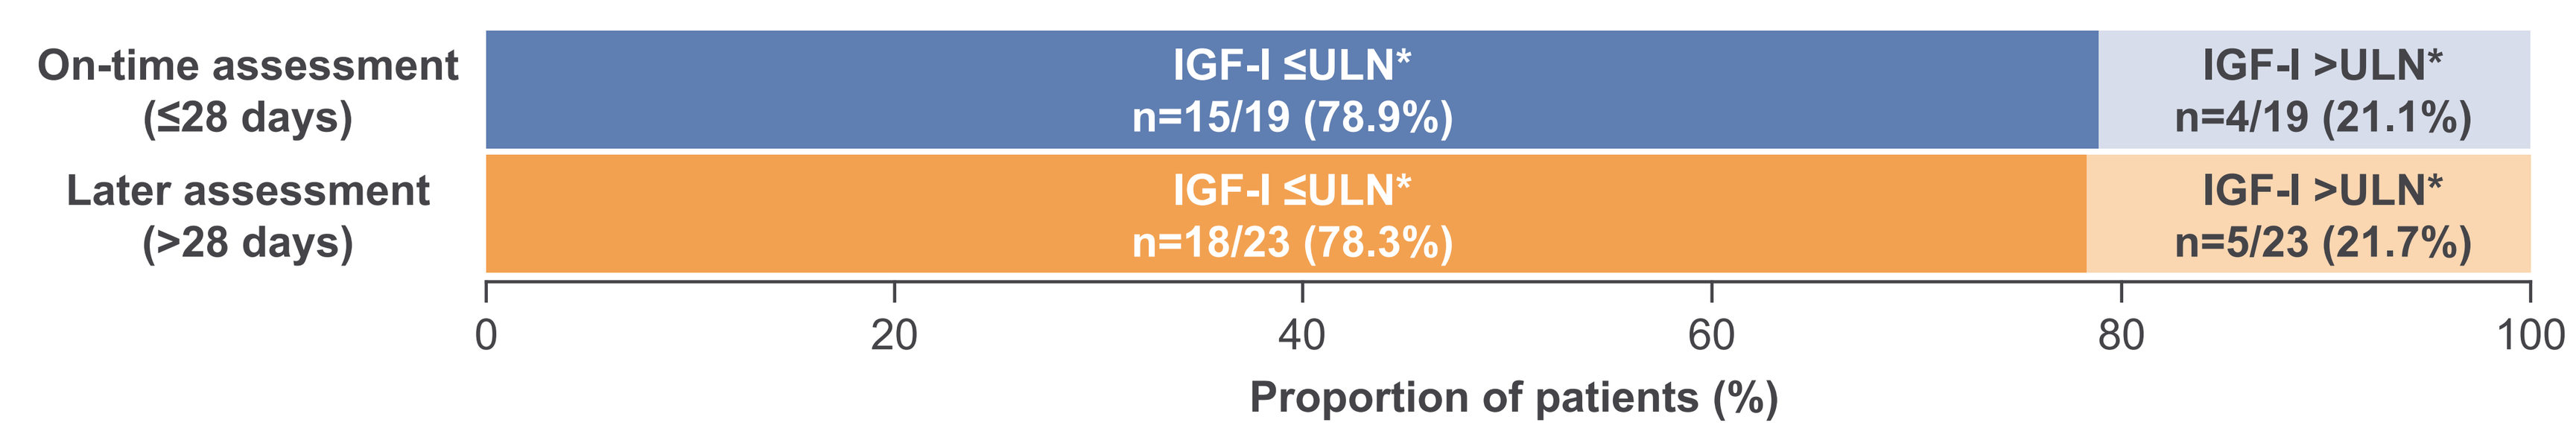
On-time assessment (≤28 days)
IGF-I ≤ULN* n=15/19 (78.9%)
IGF-I >ULN* n=4/19 (21.1%)
Later assessment (>28 days)
IGF-I ≤ULN* n=18/23 (78.3%)
IGF-I >ULN* n=5/23 (21.7%)
0 20 40 60 80 100
Proportion of patients (%)

Data are for patients who completed their W20 dose and post-final dose assessments (N=42). *ULN per age and sex.

Mean IGF-I values remained stable throughout the dose-to-assessment interval, including in patients with assessments performed >28 days post-dose

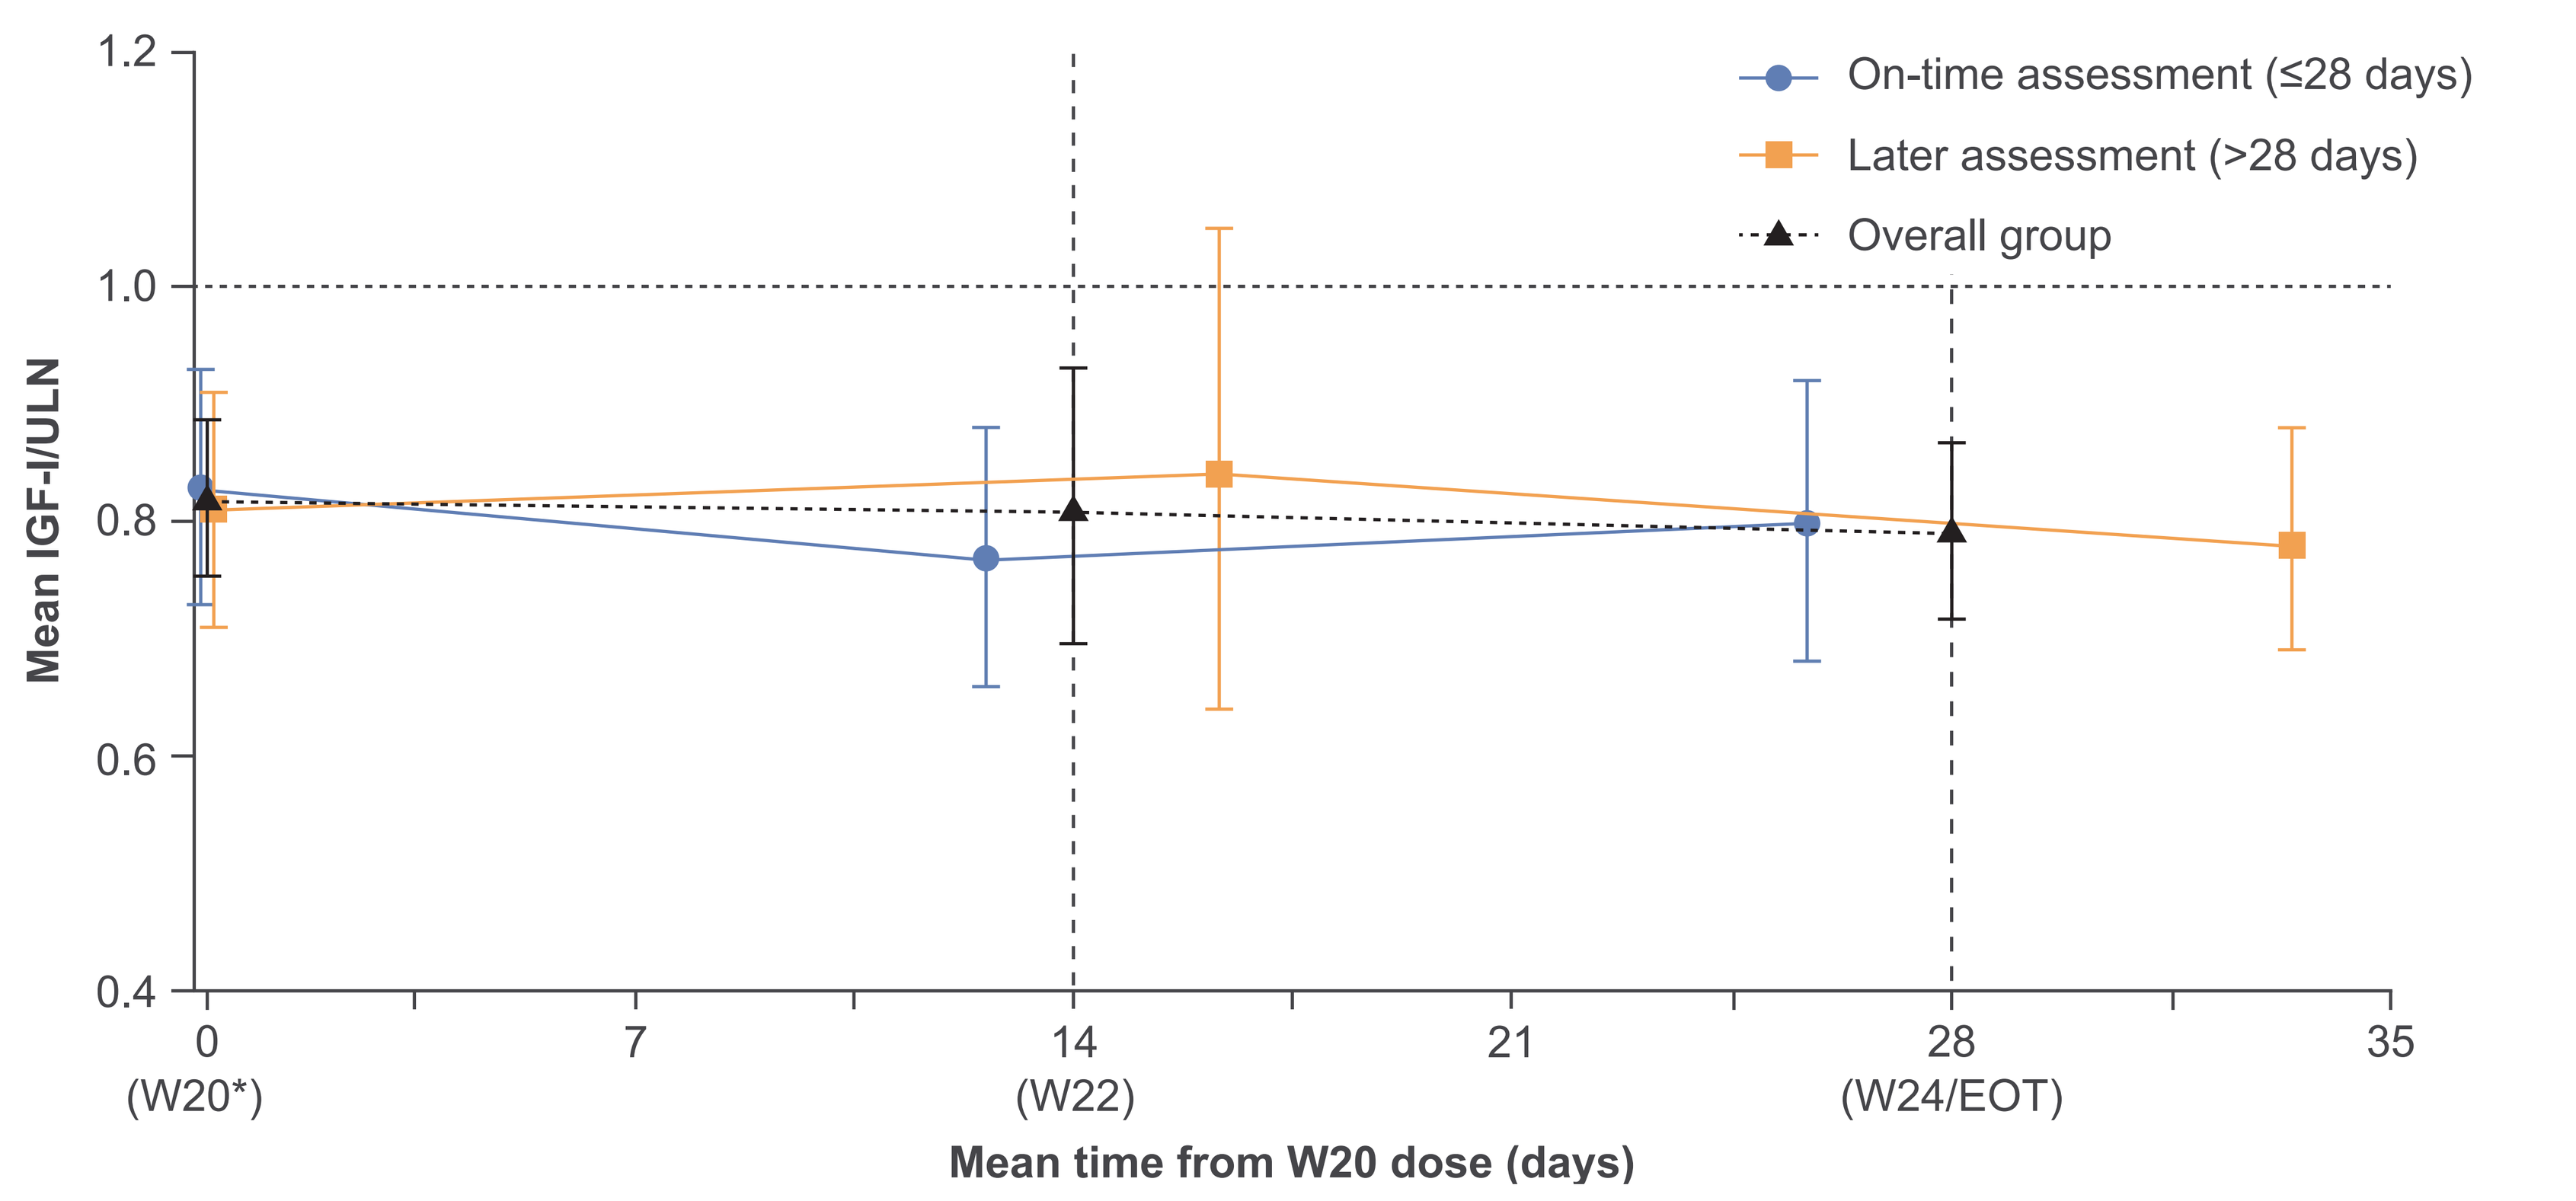
On-time assessment (≤28 days)
Later assessment (>28 days)
Overall group
Mean IGF-I/ULN
1.2
1.0
0.8
0.6
0.4
0 (W20*)
7
14 (W22)
21
28 (W24/EOT)
35
Mean time from W20 dose (days)

*W20 assessments were conducted prior to CAM2029 administration. Data are for patients who completed their W20 dose and post-final dose assessments (on-time assessments at W20, W22 and W24/EOT, n=19; later assessments at W20 and W24/EOT, n=23; later assessments at W22, n=22). Error bars represent ±95% CI. The grey horizontal rule represents ULN per age and sex. CI, confidence interval.

IGF-I values generally remained stable in individual patients, including in those with assessments performed >28 days post-dose

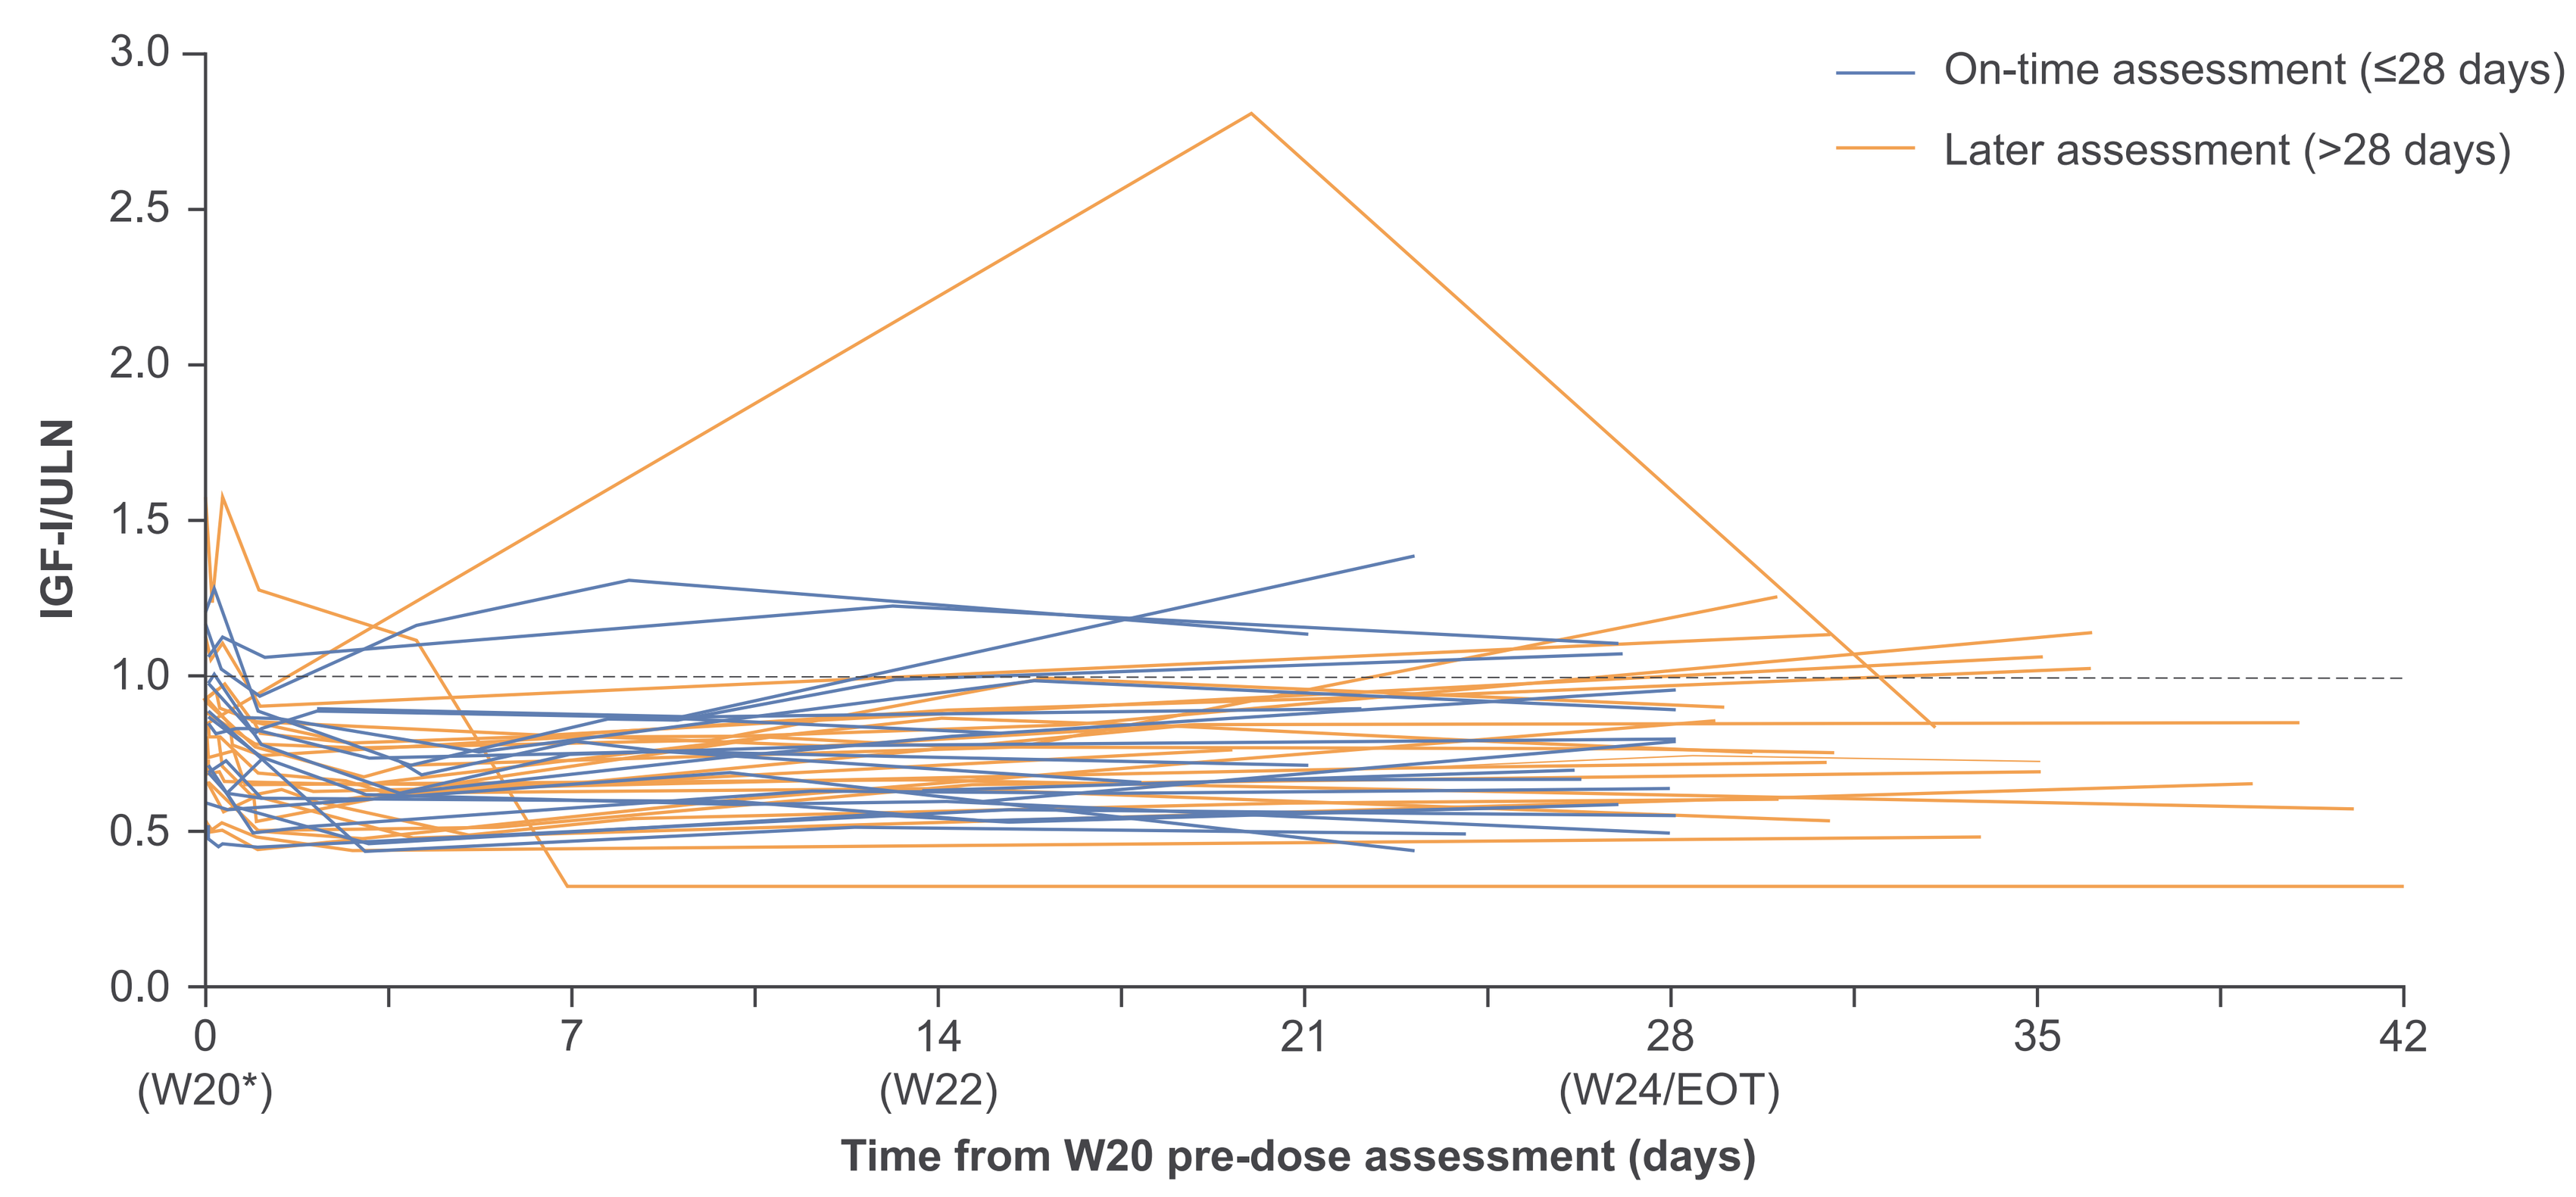
On-time assessment (≤28 days)
Later assessment (>28 days)
IGF-I/ULN
3.0
2.5
2.0
1.5
1.0
0.5
0.0
0 (W20*)
7
14 (W22)
21
28 (W24/EOT)
35
42
Time from W20 pre-dose assessment (days)

*W20 assessments were conducted prior to CAM2029 administration. Additional samples taken at 2±1 hours, 5±1 hours, 8±1 hours, 24±4 hours and 96±24 hours post-CAM2029 administration are shown here. Data are for patients who completed their W20 dose and post-final dose assessments (on-time assessments at W20, W22 and W24/EOT, n=19; later assessments at W20 and W24/EOT, n=23; later assessments at W22, n=22). The grey horizontal rule represents ULN per age and sex. Results for the W22 assessment show a single outlier result with raised IGF-I; this patient had normal IGF-I levels at W20 and W24/EOT assessments, suggesting this result is an artefact.

AIS overall scores were stable throughout the dose-to-assessment interval, including in patients with assessments performed >28 days post-dose

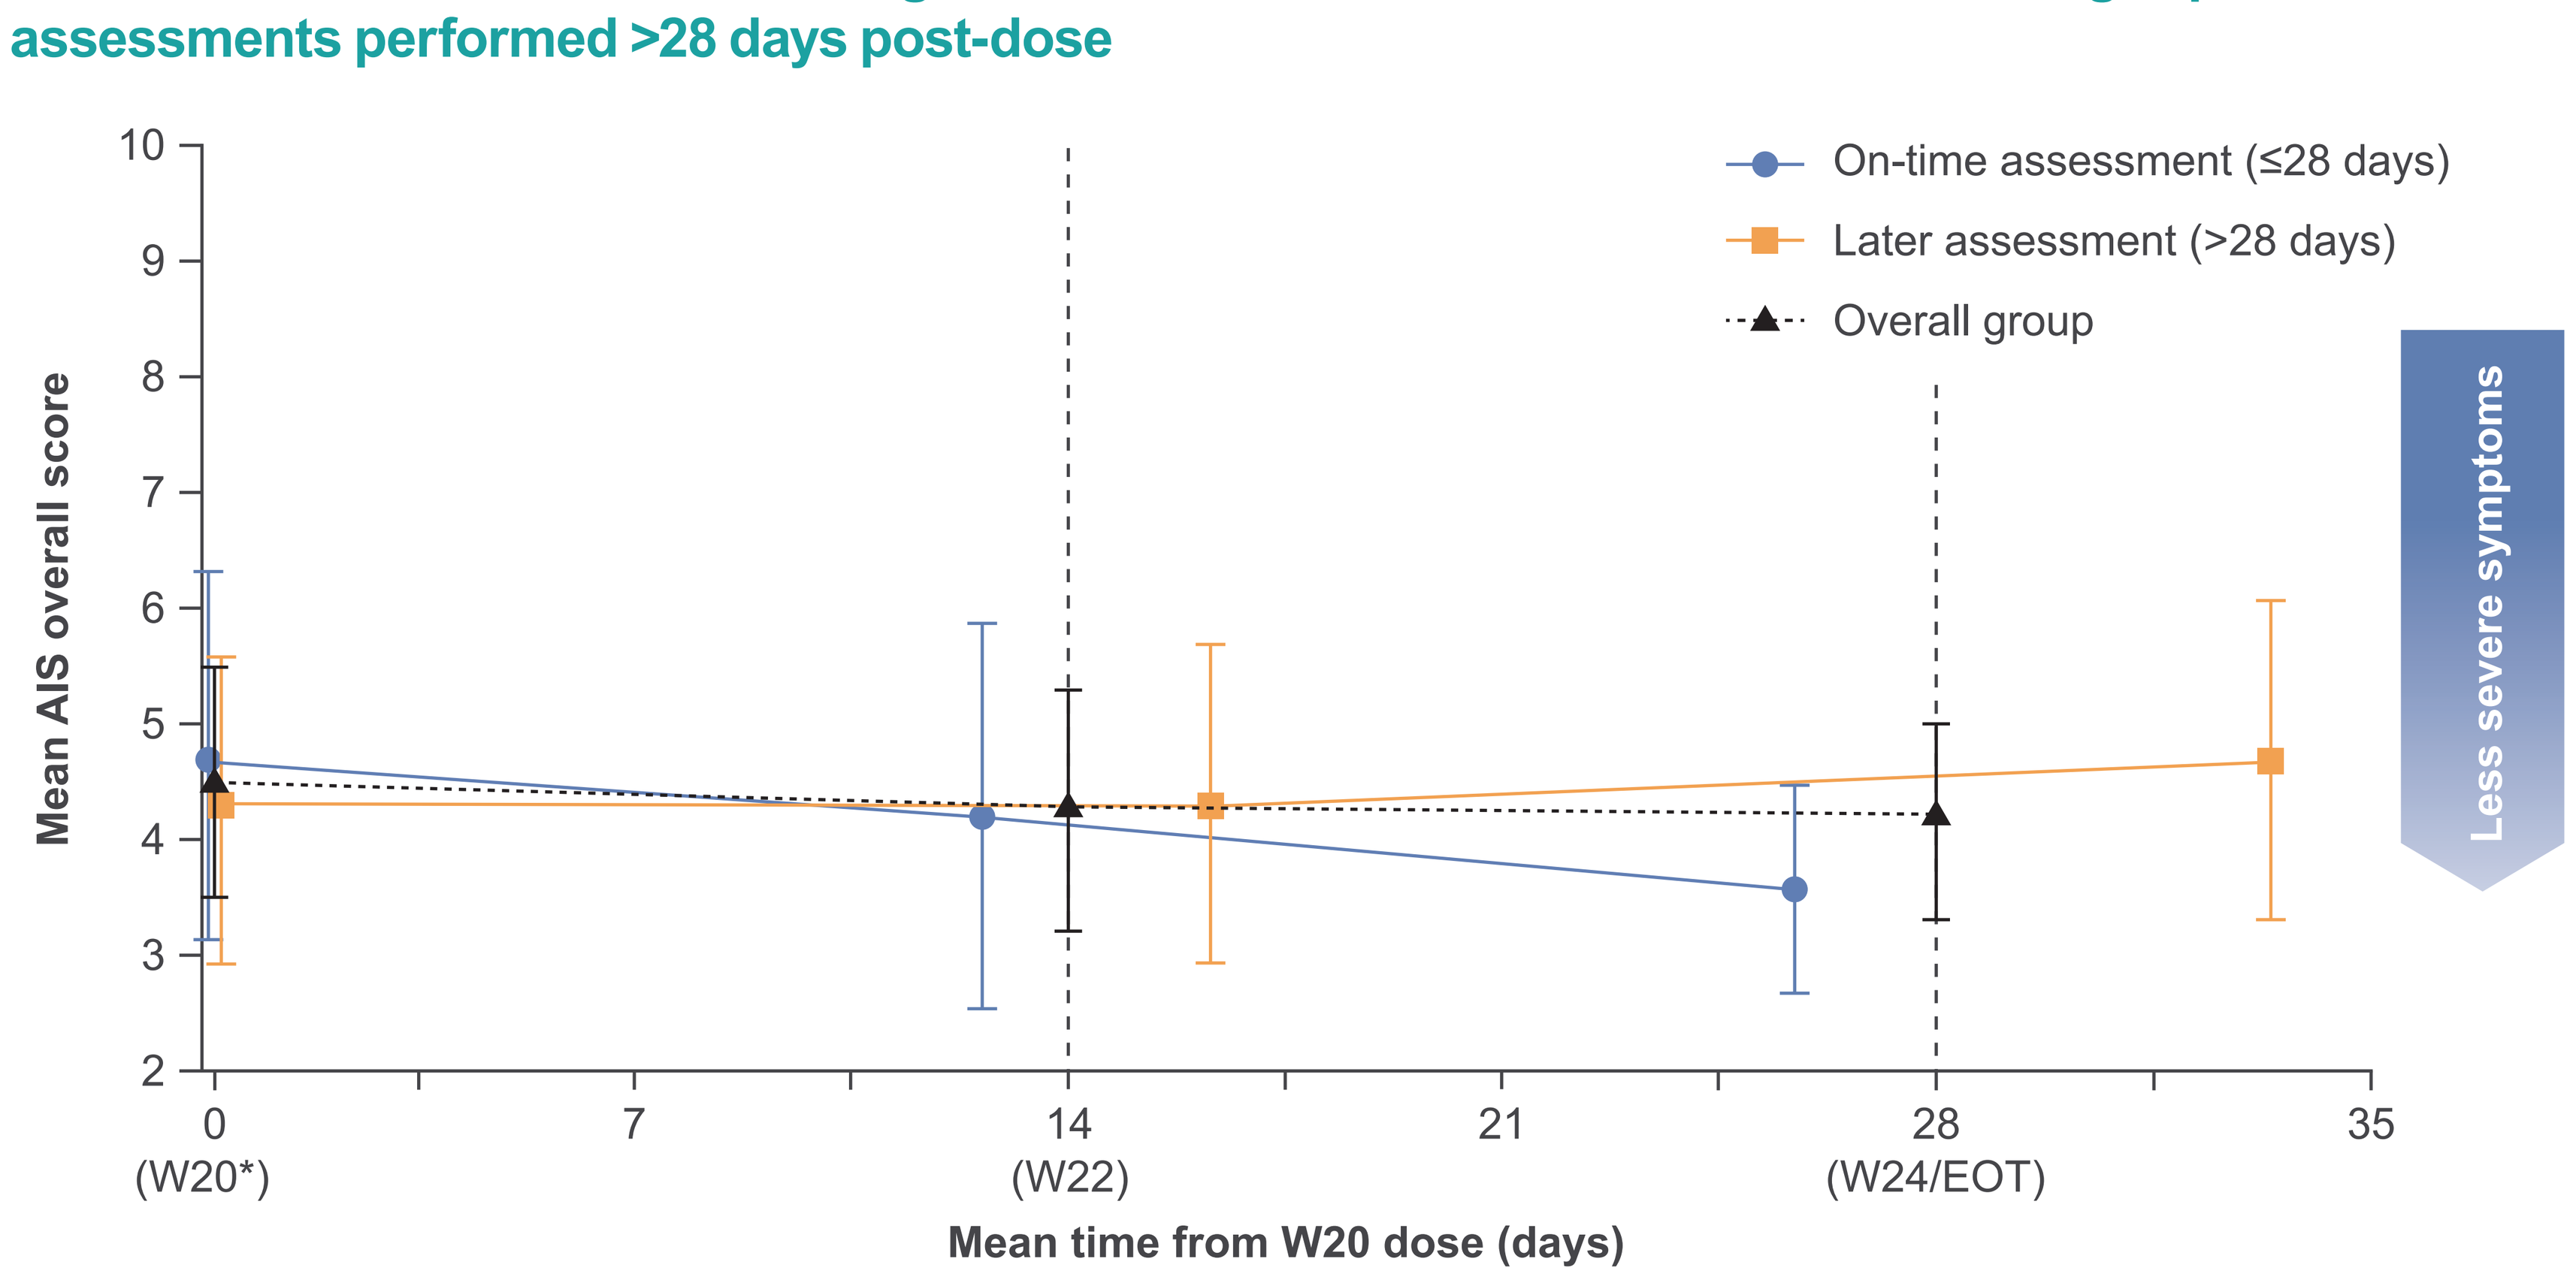
On-time assessment (≤28 days)
Later assessment (>28 days)
Overall group
Less severe symptoms
Mean AIS overall score
10
9
8
7
6
5
4
3
2
0 (W20*)
7
14 (W22)
21
28 (W24/EOT)
35
Mean time from W20 dose (days)

*W20 assessments were conducted prior to CAM2029 administration. Data are for patients who completed their W20 dose and post-final dose assessments (on-time assessments at W20, W22 and W24/EOT, n=19; later assessments at W20 and W24/EOT, n=23; later assessments at W22, n=22). AIS scores range from 0 (lowest) to 18 (highest); sum of 6 scores (0–3; none–severe) for headache, sweating, fatigue, joint pain, paraesthesia, and soft tissue swelling. A reduction in AIS overall score indicates improvement in symptoms. Error bars represent ±95% CI. Individual patient AIS scores were generally stable, including among those with later assessments. Control of individual symptoms was maintained throughout the dose-to-assessment interval, including among patients with assessments performed >28 days post-dose. See **Supplementary Figure 3** for more details, available via the QR code.

AIS overall scores were generally stable in individual patients, including in those with assessments performed >28 days post-dose

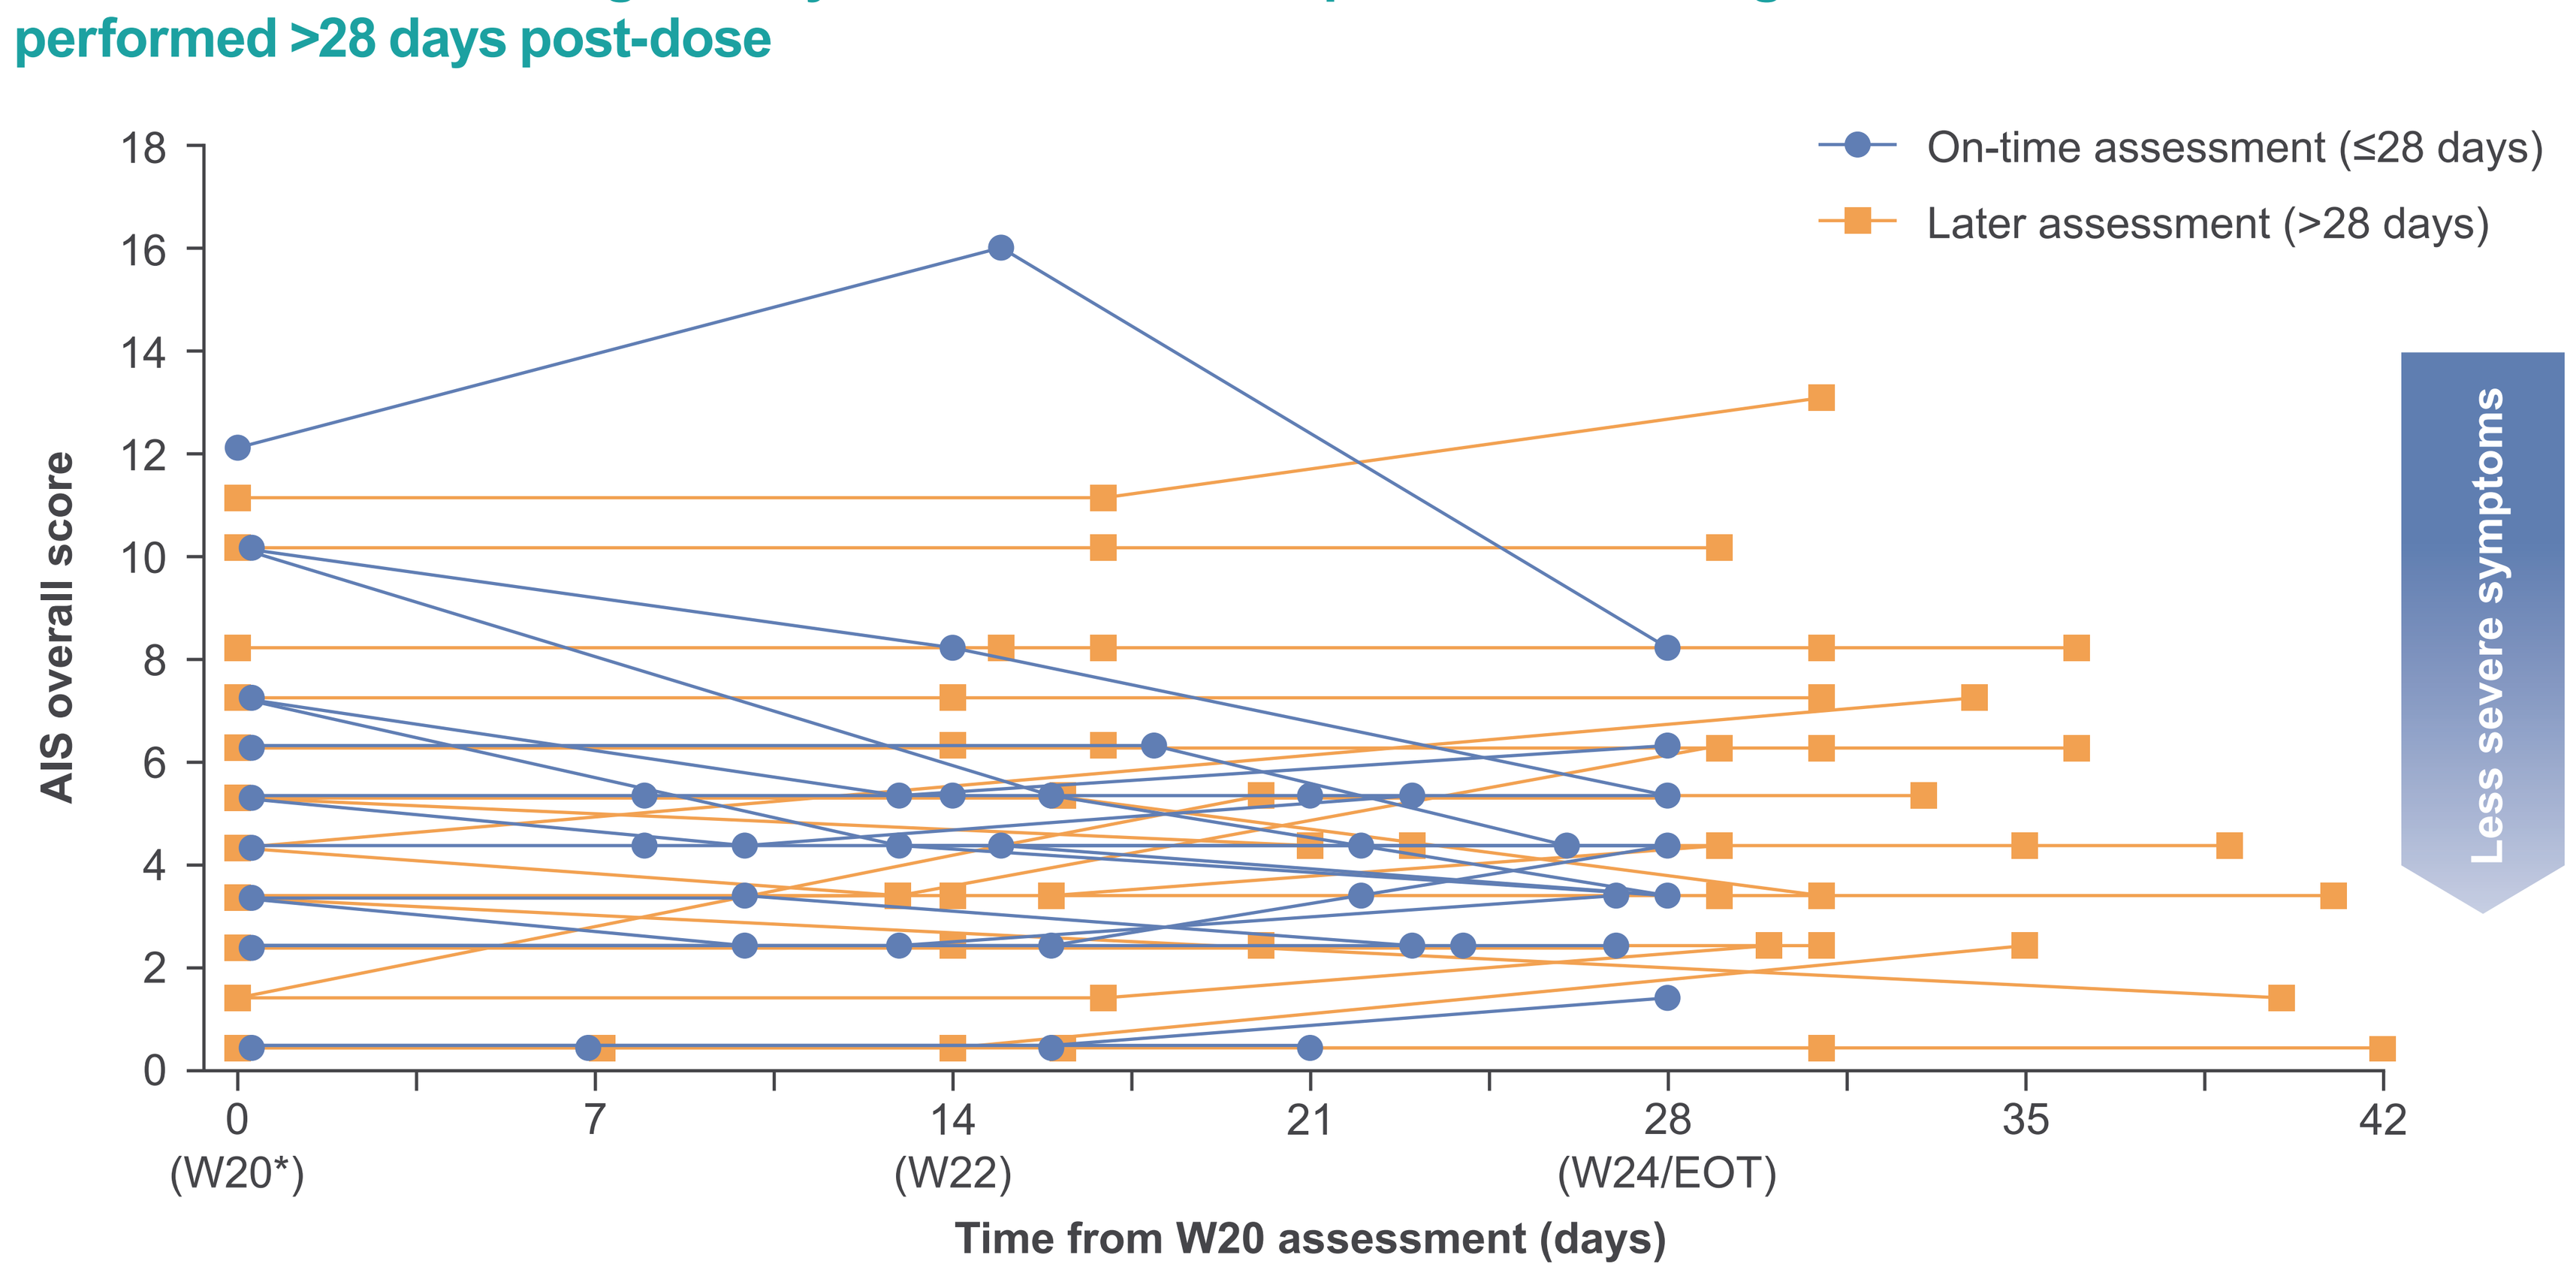
On-time assessment (≤28 days)
Later assessment (>28 days)
Less severe symptoms
AIS overall score
18
16
14
12
10
8
6
4
2
0
0 (W20*)
7
14 (W22)
21
28 (W24/EOT)
35
42
Time from W20 assessment (days)

*W20 assessments were conducted prior to CAM2029 administration. Data are for patients who completed their W20 dose and post-final dose assessments (on-time assessments at W20, W22 and W24/EOT, n=19; later assessments at W20 and W24/EOT, n=23; later assessments at W22, n=22). AIS scores range from 0 (lowest) to 18 (highest); sum of 6 scores (0–3; none–severe) for headache, sweating, fatigue, joint pain, paraesthesia, and soft tissue swelling. A reduction in AIS overall score indicates improvement in symptoms.

References

1. Gadelha MR *et al. Endocr Rev* 2019;40:268–332; **2.** Giustina A *et al. Pituitary* 2024;27:7–22; **3.** Giustina A *et al. Rev Endocr Metab Disord* 2020;21:667–78; **4**. Fleseriu M *et al. Lancet Diabetes Endocrinol* 2022;10:804–26; **5.** Gurel MH *et al. Patient Prefer Adherence* 2014;8:53–62; **6.** Strasburger CJ *et al. Eur J Endocrinol* 2016;174:355–62; **7**. Geer EB *et al. BMC Endocr Disord* 2020;20:117; **8**. Tiberg F *et al. Br J Clin Pharmacol* 2015;80:460–72; **9.** Ferone D *et al. J Clin Endocrinol Metab* 2025;110:1729–39.

Acknowledgements and disclosures

The ACROINNOVA 1 trial was funded by Camurus AB. Thank you to the patients, investigators, nurses and trial coordinators who made this trial possible. Medical writing assistance was provided by Lisa Heaney, PhD, at Amiculum, and was funded by Camurus AB.

COI disclosures: AMP is employed by Camurus AB (CAB), was previously employed by Recordati Rare Diseases (RRD), and holds stock in CAB, Novartis and RRD. MRG has served as an advisor to CAB, Crinetics Pharmaceuticals (CP), RRD and Novo Nordisk (NN); received fees from CP, CAB, RRD and NN; and received research funding from CP and RRD. BMKB received occasional consulting honoraria from Ascendis, CAB, Chiesi, CP, Lundbeck, Neurocrine, NN, Pfizer, RRD and Xeris and received institutional research grants for clinical trials from Ascendis, NN and CP. MF received fees from AP, CAB, CP, DebioPharm and RRD and received research funding from AP, CP and DebioPharm. JLSS has served as an advisor to CAB, CP and Corcept Therapeutics; received fees from CAB; and received research funding from CAB, RRD and Alexion Pharmaceuticals (AP). JMS received fees from Xeris and CAB and received research funding from DebioPharm, Xeris, Lundbeck, NN and AbbVie. JR and FT are employed by CAB and hold stock in CAB. IB and DF have nothing to declare.

Presented at the 22nd International Congress of Endocrinology (ICE); Kyoto, Japan; 2–5 June 2026.

Previously presented at the Joint Congress of the European Society for Paediatric Endocrinology and the European Society of Endocrinology (ESPE/ESE); Copenhagen, Denmark; 10–13 May 2025, the Endocrine Society's Annual Meeting (ENDO); San Francisco, CA, USA; 12–15 July 2025, the Deutsche Gesellschaft für Endokrinologie (DGE) Herbstsitzung der AG Hypophyse und 28. Jahrestagung der Sektion Neuroendokrinologie (Joint Meeting); Hamburg, Germany; 9–10 October 2025 and the Workshop of the European Neuroendocrine Association (ENEA) Marseille, France; 3–5 December 2025.