Poster no.
PA3-N-004

Patient-reported outcomes following CAM2029 treatment for patients with acromegaly in the Phase 3 ACROINNOVA 2 trial

Diego Ferone,[1] Beverly M. K. Biller,[2] Mônica R. Gadelha,[3] Julie M. Silverstein,[4] Pinar Kadioğlu,[5] Jochen Seufert,[6] Maria Fleseriu,[7] Alberto M. Pedroncelli,[8] Jacob Råstam,[8] Maria Harrie,[8] Agneta Svedberg,[8] Fredrik Tiberg,[8] Joanna L. Spencer-Segal[9]

[1]Endocrinology Unit, Department of Internal Medicine, IRCCS Ospedale Policlinico San Martino, Genova, Italy; [2]Neuroendocrine and Pituitary Tumor Clinical Center, Massachusetts General Hospital, Boston, MA, USA; [3]Neuroendocrinology Research Center/Endocrinology Division, Medical School and Hospital Universitario Clementino Fraga Filho, Universidade Federal do Rio de Janeiro, Rio de Janeiro, Brazil; [4]Division of Endocrinology, Metabolism and Lipid Research, Department of Neurosurgery, Washington University School of Medicine, St. Louis, MO, USA; [5]Division of Endocrinology-Metabolism and Diabetes, Department of Internal Medicine, Istanbul University-Cerrahpaşa, Istanbul, Turkey; [6]Division of Endocrinology and Diabetology, Department of Medicine II, Medical Center, University of Freiburg, Freiburg, Germany; [7]Pituitary Center, Departments of Medicine and Neurological Surgery, Oregon Health and Science University, Portland, OR, USA; [8]Camurus AB, Lund, Sweden; [9]Department of Internal Medicine and Michigan Neuroscience Institute, University of Michigan, Ann Arbor, MI, USA

Scan QR code
for poster and supplement

All materials obtained through the QR code are for personal use only and may not be reproduced without permisson of the authors

Presenting author: Alberto M. Pedroncelli

BACKGROUND

- For patients with acromegaly, the ability to self-administer medication and the associated ease of administration are important[1]
- Injectable first-generation somatostatin receptor ligands (SRLs; octreotide long-acting repeatable [LAR] or lanreotide Autogel [ATG]) can provide effective biochemical control but are typically administered by a healthcare professional and can result in a substantial treatment burden[2–6]
- There is an unmet need for therapies that can provide biochemical control and reduce symptoms, have a lower treatment burden and do not require administration by healthcare professionals[6,7]
- CAM2029 is a novel octreotide subcutaneous depot (based on FluidCrystal® technology) with a long-acting formula for convenient monthly self-administration via a ready-to-use autoinjector pen with small-gauge needle[8,9] (see **Supplementary Figure 1**, available via the QR code)
- In ACROINNOVA 1 (NCT04076462), a 24-week, Phase 3, randomised, double-blind trial, CAM2029 achieved superior biochemical control versus placebo (72.2% vs 37.5%, respectively; *P*=0.0018), and improved quality of life (QoL) in patients with insulin-like growth factor I (IGF-I) ≤ upper limit of normal per age and sex (ULN) receiving standard-of-care (SoC) treatment (octreotide LAR/lanreotide ATG) at screening[8]
- ACROINNOVA 2 (NCT04125836) was a 52-week, Phase 3, open-label trial in which the long-term safety and efficacy of CAM2029 were assessed in patients with acromegaly who had previously received first-generation SRLs (current SoC); it was complemented by a 52-week extension phase
 - Patient-reported outcomes (PROs) for the complete data set from the core phase of the trial are reported here; for corresponding safety and biochemical control analyses, see poster PA2-N-036

CONCLUSIONS

CAM2029 showed a long-term safety profile similar to SoC

Long-term CAM2029 treatment continuously improved QoL and treatment satisfaction versus SoC baseline, as measured by multiple PRO instruments

ACROINNOVA 2 demonstrates the combined benefits of convenient administration, improved QoL and treatment satisfaction provided by CAM2029

METHODS

Analyses for the core phase of ACROINNOVA 2 were undertaken at week 52

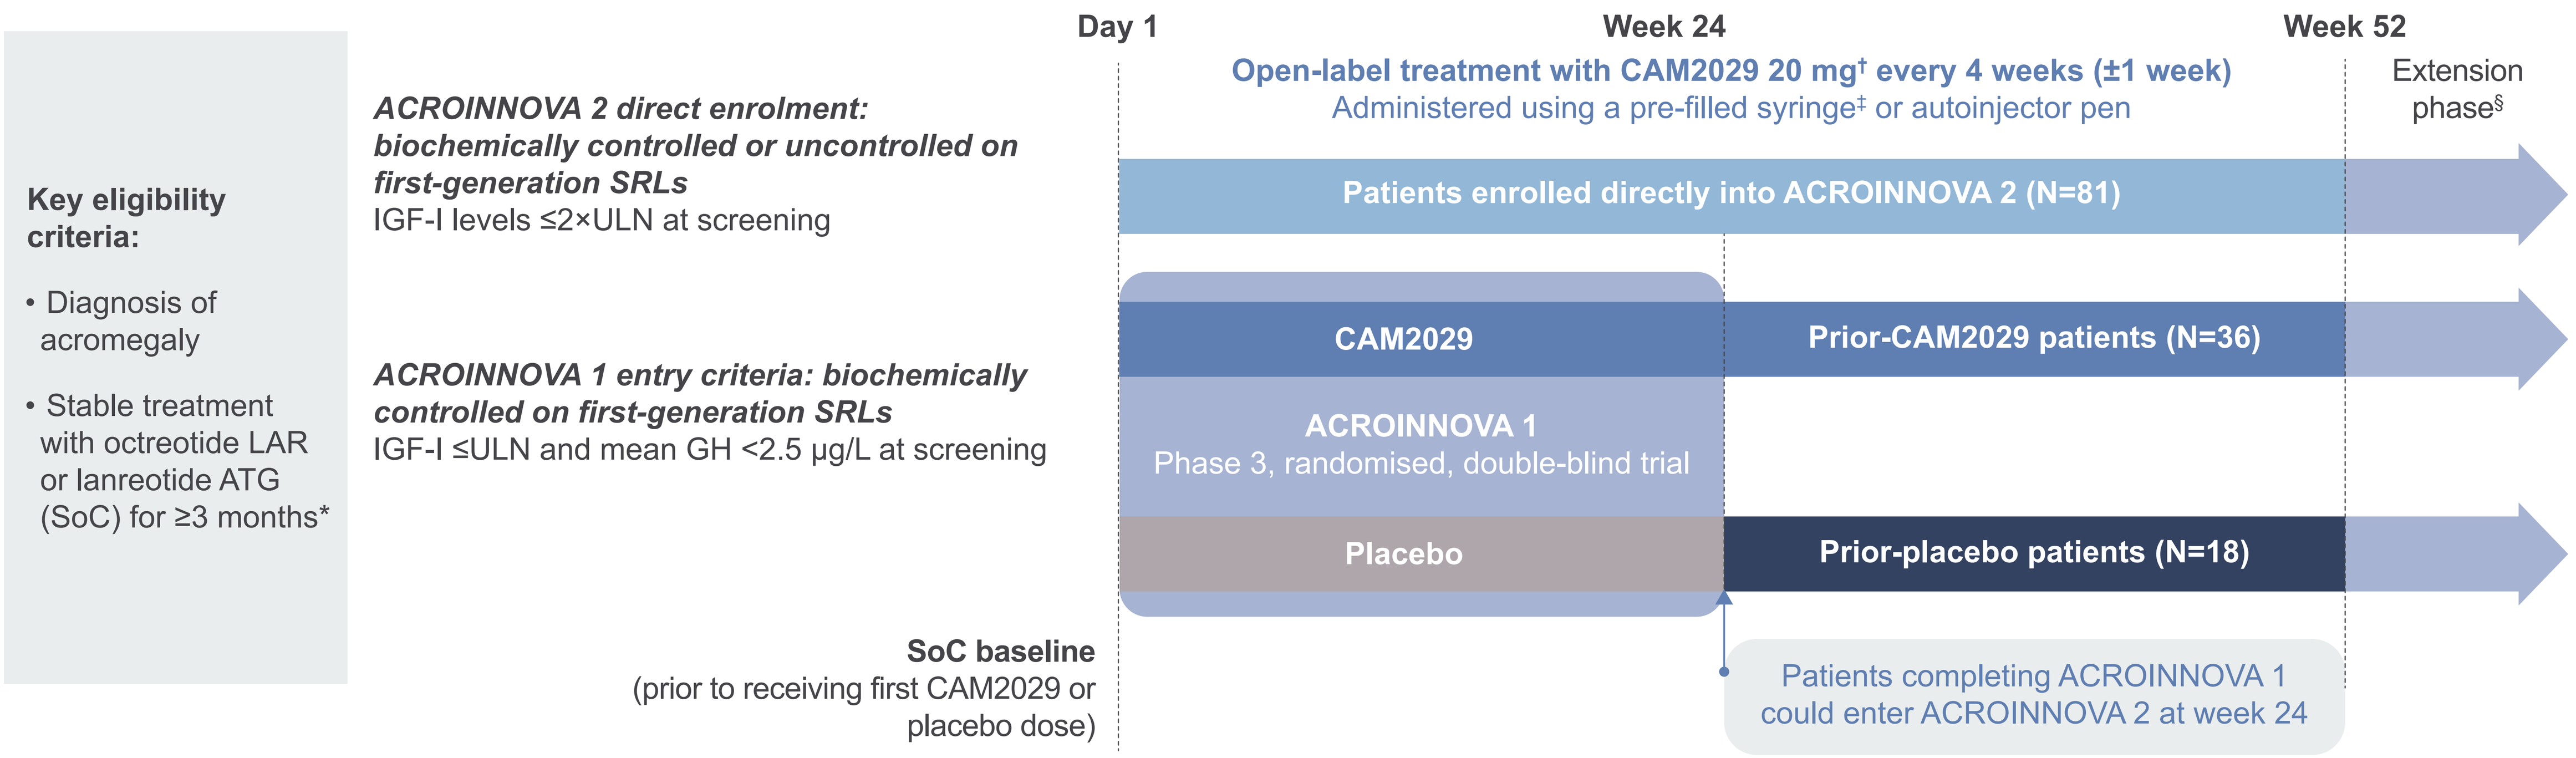

Key eligibility criteria:
• Diagnosis of acromegaly
• Stable treatment with octreotide LAR or lanreotide ATG (SoC) for ≥3 months*
ACROINNOVA 2 direct enrolment: biochemically controlled or uncontrolled on first-generation SRLs
IGF-I levels ≤2×ULN at screening
ACROINNOVA 1 entry criteria: biochemically controlled on first-generation SRLs
IGF-I ≤ULN and mean GH <2.5 µg/L at screening
Day 1
Week 24
Week 52
Open-label treatment with CAM2029 20 mg† every 4 weeks (±1 week)
Administered using a pre-filled syringe‡ or autoinjector pen
Extension phase§
Patients enrolled directly into ACROINNOVA 2 (N=81)
CAM2029
Prior-CAM2029 patients (N=36)
ACROINNOVA 1
Phase 3, randomised, double-blind trial
Placebo
Prior-placebo patients (N=18)
SoC baseline
(prior to receiving first CAM2029 or placebo dose)
Patients completing ACROINNOVA 1 could enter ACROINNOVA 2 at week 24

The overall population includes the prior-placebo group who received 24 weeks of placebo before switching to CAM2029. *Treatment with a stable dose of octreotide LAR (10, 20, 30 or 40 mg) or lanreotide ATG (60, 90 or 120 mg); †If required based on safety and tolerability, CAM2029 dose could be reduced to 10 mg every 4 weeks (±1 week); ‡Only the autoinjector pen is available following marketing authorisation;[10] §Upon completing the core phase in ACROINNOVA 2, patients could continue to receive CAM2029 in a 52-week extension phase. GH, growth hormone.

Primary endpoint	
• Characterisation of adverse events	
Selected secondary endpoints	
PROs: QoL	• **AcroQoL:** disease-specific scale to assess QoL in patients with acromegaly*
PROs: patient satisfaction	• **TSQM:** patients' satisfaction with medication*
	• **SIAQ:** patients' overall experience with SC self-injection*
	• **PSS:** patients' overall experience with trial treatment versus baseline SoC*

*Details for each PRO (background, measures, scoring) are provided in **Supplementary Figure 2**, available via the QR code. AcroQoL, Acromegaly Quality of Life Questionnaire; PSS, Patient Satisfaction Scale; SIAQ, Self-Injection Assessment Questionnaire; TSQM, Treatment Satisfaction Questionnaire for Medication.

RESULTS

- Of 135 patients enrolled in ACROINNOVA 2, 127 (94%) completed treatment
- Mean (standard deviation [SD]) age was 52.9 (11.9) years, 56.3% of patients were female and 88.1% had undergone prior pituitary surgery (see poster PA2-N-036)
- Most (91.1%) patients (or partners) opted to self-administer CAM2029
- CAM2029 was well tolerated, with no new safety signals up to week 52 (see **Supplementary Table 1**, available via QR code, and poster PA2-N-036)

In the overall population, AcroQoL* scores improved from baseline to week 52

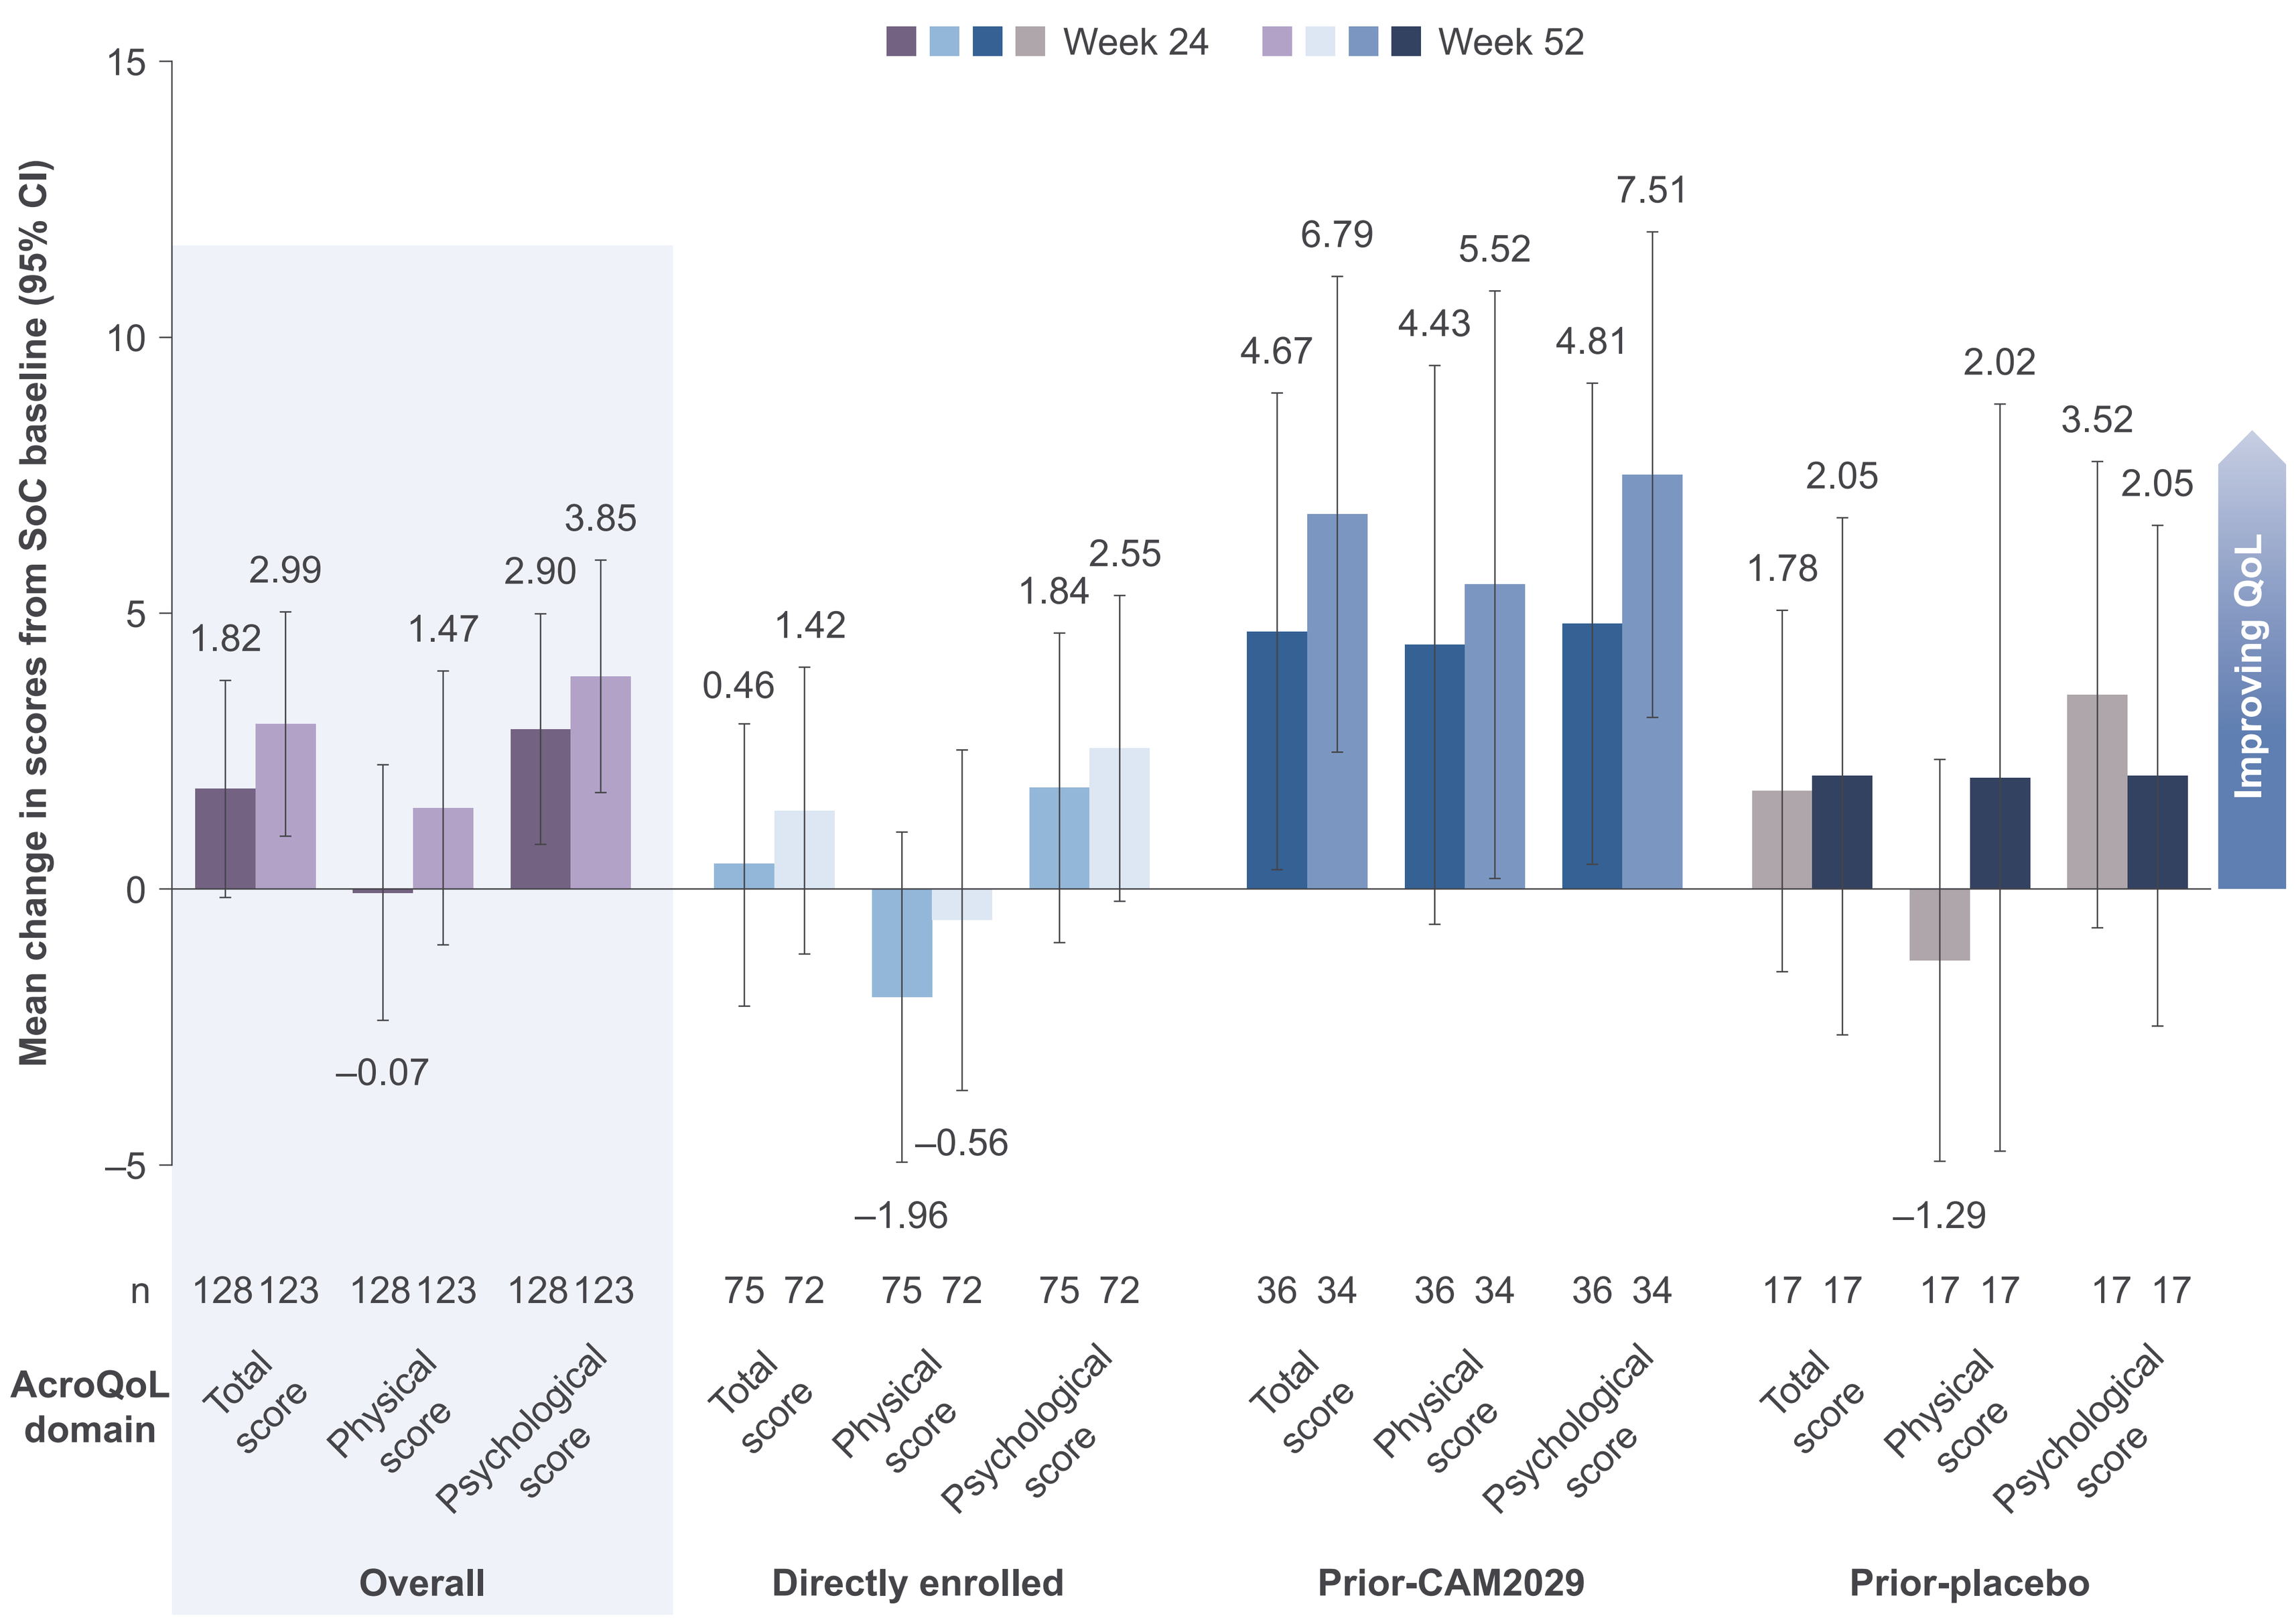

Week 24
Week 52
Mean change in scores from SoC baseline (95% CI)
15
10
5
0
–5
1.82
2.99
–0.07
1.47
2.90
3.85
0.46
1.42
–1.96
–0.56
1.84
2.55
4.67
6.79
4.43
5.52
4.81
7.51
1.78
2.05
–1.29
2.02
3.52
2.05
Improving QoL
n
128 123
128 123
128 123
75 72
75 72
75 72
36 34
36 34
36 34
17 17
17 17
17 17
AcroQoL domain
Total score
Physical score
Psychological score
Overall
Directly enrolled
Prior-CAM2029
Prior-placebo

Data include the 24-week placebo period before the prior-placebo group switched to CAM2029; the group is also included in the overall population. *Score range: 0–100.[11] CI, confidence interval.

- At week 52 in the treatment subgroups, AcroQoL scores were numerically improved versus SoC baseline, except for physical score in the directly enrolled group
 - AcroQoL scores were also numerically greater at week 52 than week 24, indicating continued improvements, except for psychological score in the prior-placebo group

In the overall population, TSQM* scores improved from baseline to week 52

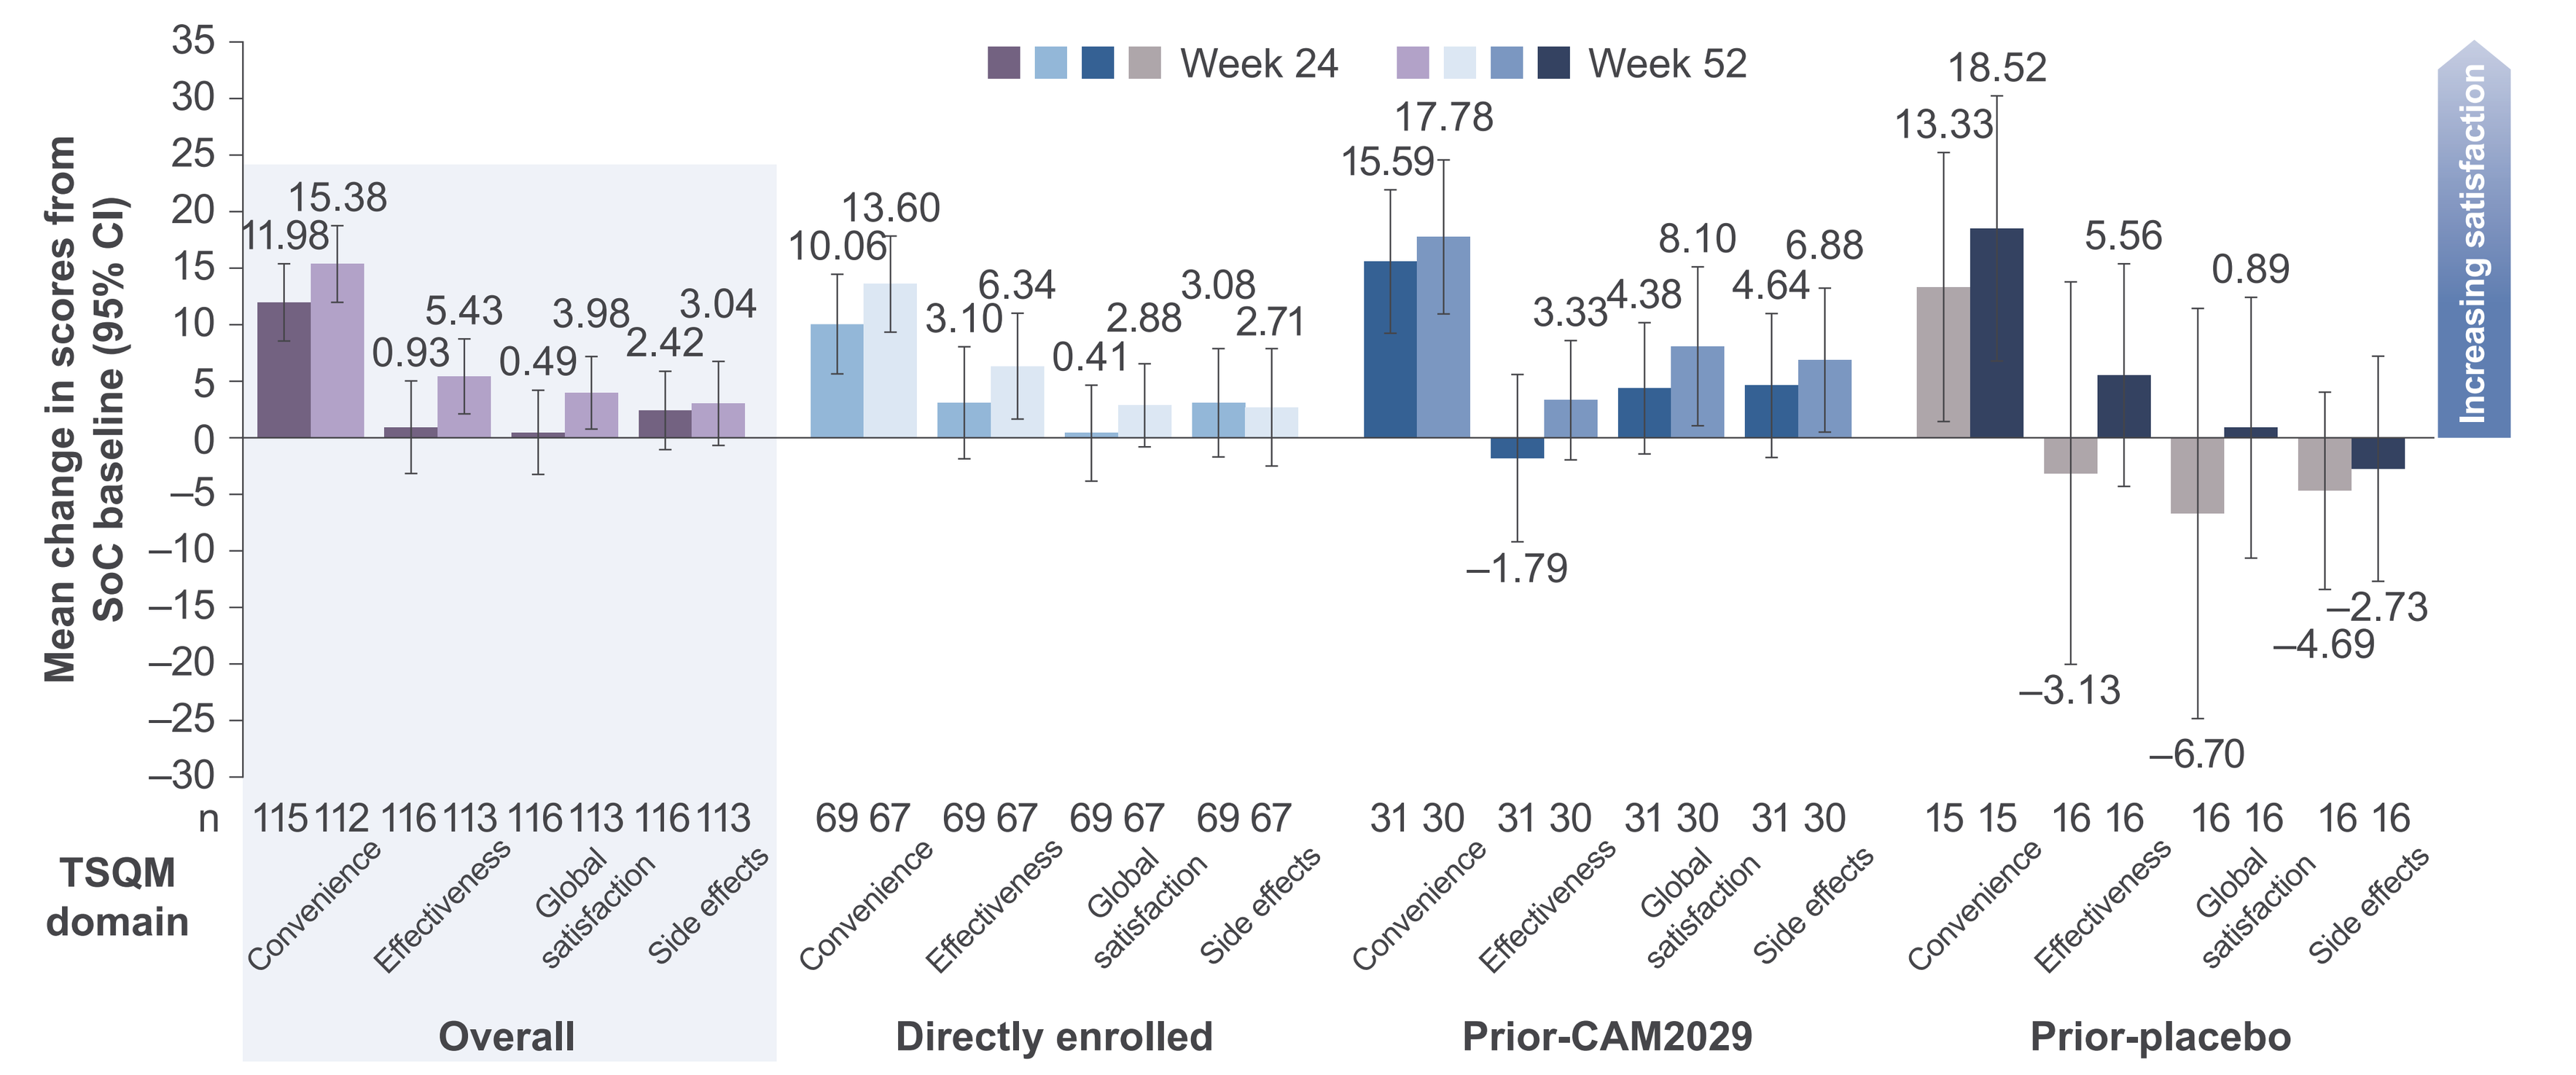

Week 24
Week 52
Mean change in scores from SoC baseline (95% CI)
35
30
25
20
15
10
5
0
–5
–10
–15
–20
–25
–30
11.98
15.38
0.93
5.43
0.49
3.98
2.42
3.04
10.06
13.60
3.10
6.34
0.41
2.88
3.08
2.71
15.59
17.78
–1.79
3.33
4.38
8.10
4.64
6.88
13.33
18.52
–3.13
5.56
–6.70
0.89
–4.69
–2.73
Increasing satisfaction
n
115 112
116 113
116 113
116 113
69 67
69 67
69 67
69 67
31 30
31 30
31 30
31 30
15 15
16 16
16 16
16 16
TSQM domain
Convenience
Effectiveness
Global satisfaction
Side effects
Overall
Directly enrolled
Prior-CAM2029
Prior-placebo

Data include the 24-week placebo period before the prior-placebo group switched to CAM2029; the group is also included in the overall population. *Score range: 0–100.[12]

- TSQM scores were numerically improved (week 52 vs SoC baseline), except for side effects in the prior-placebo group
 - TSQM scores were also numerically greater at week 52 than week 24, indicating continued improvements, except for side effects in the directly enrolled group

In the overall population, SIAQ* scores and treatment satisfaction improved from baseline and were maintained long term

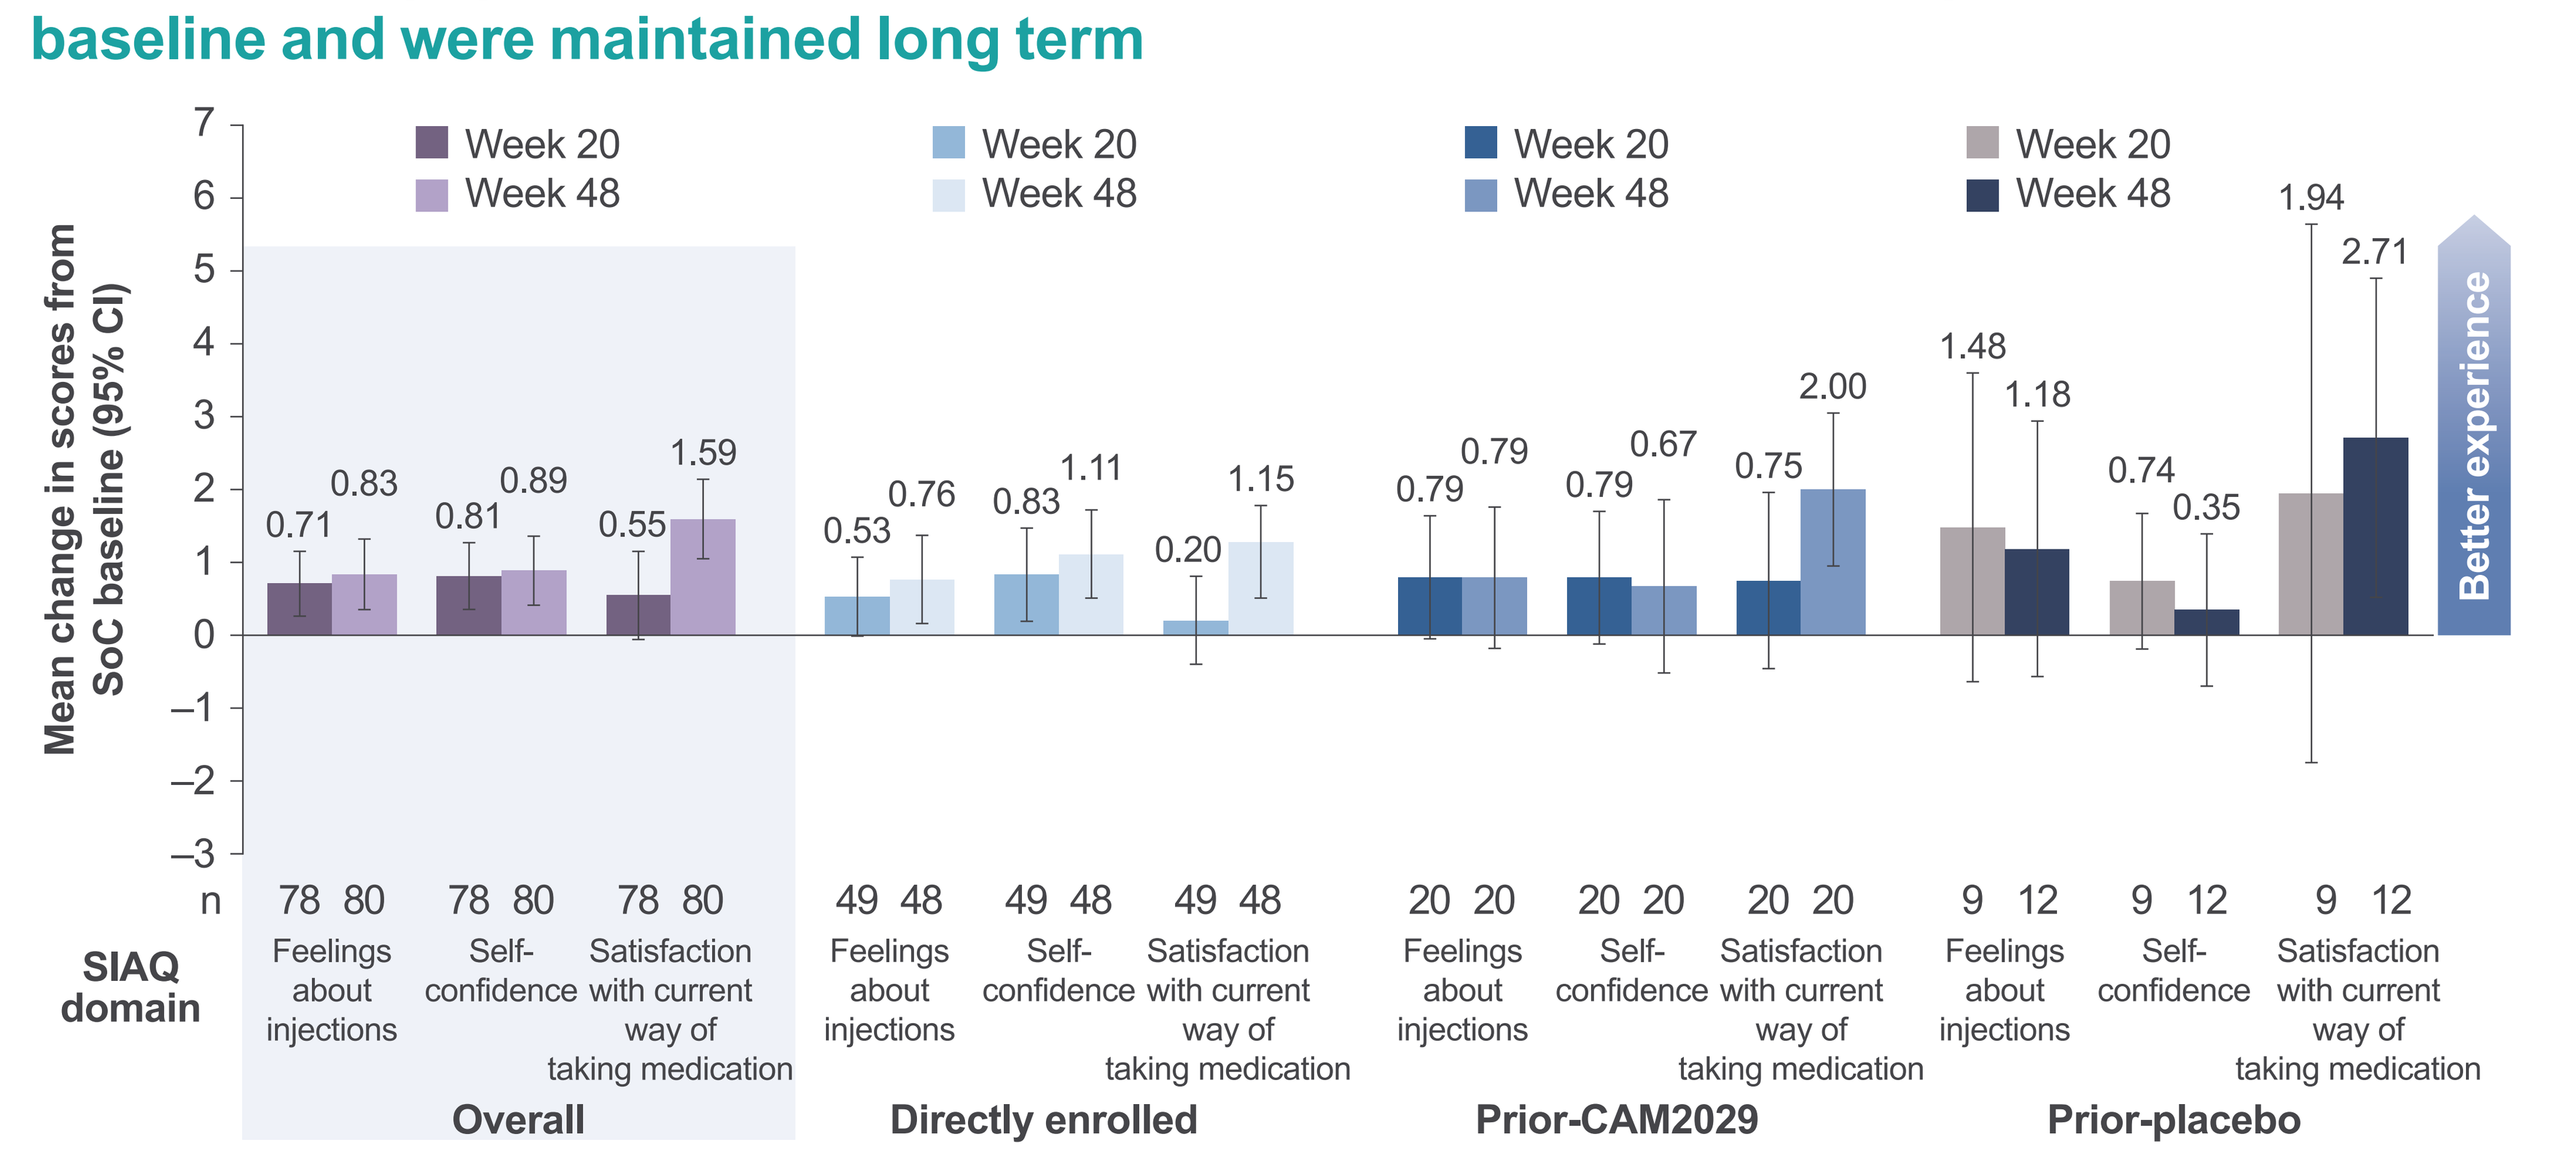

Week 20
Week 48
Mean change in scores from SoC baseline (95% CI)
7
6
5
4
3
2
1
0
–1
–2
–3
0.71
0.83
0.81
0.89
0.55
1.59
0.53
0.76
0.83
1.11
0.20
1.15
0.79
0.79
0.79
0.67
0.75
2.00
1.48
1.18
0.74
0.35
1.94
2.71
Better experience
n
78 80
78 80
78 80
49 48
49 48
49 48
20 20
20 20
20 20
9 12
9 12
9 12
SIAQ domain
Feelings about injections
Self-confidence
Satisfaction with current way of taking medication
Overall
Directly enrolled
Prior-CAM2029
Prior-placebo

Patients assessed at week 48 or closest visit. Data include the 24-week placebo period before the prior-placebo group switched to CAM2029; the group is also included in the overall population. *Score range: 0–10.[13]

- At week 52, a high proportion of patients rated their experience with CAM2029 as 'much better' (41.5%) or 'slightly better' (23.0%) on the PSS, compared with baseline SoC

References

1. Cella D *et al. Adv Ther* 2021;38:969–93; **2.** Melmed S *et al. Nat Rev Endocrinol* 2018;14:552–61; **3.** Jørgensen JOL *et al. Adv Ther* 2023;40:4675–88; **4.** Geer EB *et al. BMC Endocrine Disord* 2020;20:117; **5.** Gurel MH *et al. Patient Prefer Adherence* 2014;8:53–62; **6.** Fleseriu M *et al. Front Endocrinol (Lausanne)* 2021;12:627711; **7.** Shah SN *et al. Front Endocrinol (Lausanne)* 2025;16:1516131; **8.** Ferone D *et al. J Clin Endocrinol Metab* 2024;00:1–11; **9.** Tiberg F *et al. Br J Clin Pharmacol* 2015;80:460–72; **10.** Camurus AB. Oczyesa® (CAM2029). EU summary of product characteristics, 2025; **11.** Badia X *et al. Health Qual Life Outcomes* 2004;2:13; **12.** Atkinson MJ *et al. Health Qual Life Outcomes* 2004;2:12; **13.** Keininger D, Coteur G. *Health Qual Life Outcomes* 2011;9:2.

Acknowledgements and disclosures

The ACROINNOVA 2 trial was funded by Camurus AB. Thank you to the patients, investigators, nurses and trial coordinators who made this trial possible. Medical writing assistance was provided by Lisa Heaney, PhD, at Amiculum® and was funded by Camurus AB.

COI disclosures: AMP is employed by Camurus AB (CAB), was previously employed by Recordati Rare Diseases (RRD), and holds stock in CAB, Novartis and RRD. BMKB received occasional consulting honoraria from Ascendis, CAB, Chiesi, CP, Lundbeck, Neurocrine, NN, Pfizer, RRD and Xeris and received institutional research grants for clinical trials from Ascendis, CP and NN. MRG has served as an advisor to CAB, Crinetics Pharmaceuticals (CP), RRD and Novo Nordisk (NN); received fees from CAB, CP, RRD and NN; and received research funding from CP and RRD. JMS received fees from Xeris and CAB and received research funding from DebioPharm, Xeris, Lundbeck, NN and AbbVie. JS received fees from CAB and received research funding from CAB. MF received fees from AP, CAB, CP, DebioPharm and RRD and received research funding from Alexion Pharmaceuticals (AP), CP and DebioPharm. JR, MH, AS and FT are employed by CAB; JR, MH and FT hold stock in CAB. AS has also served as an advisor to Pharma Holdings AS. JLSS has served as an advisor to CAB, CP and Corcept Therapeutics; received fees from CAB; and received research funding from CAB, RRD and AP. DF and PK have nothing to declare.

Presented at the 22nd International Congress of Endocrinology (ICE); Kyoto, Japan; 2–5 June 2026.

Previously presented at the Joint Congress of the European Society for Paediatric Endocrinology and the European Society of Endocrinology; Copenhagen, Denmark (ESPE/ESE); 10–13 May 2025, the Endocrine Society's Annual Meeting (ENDO); San Francisco, CA, USA; 12–15 July 2025, the Deutsche Gesellschaft für Endokrinologie (DGE) Herbstsitzung der AG Hypophyse und 28. Jahrestagung der Sektion Neuroendokrinologie (Joint Meeting); Hamburg, Germany; 9–10 October 2025 and the Workshop of the European Neuroendocrine Association (ENEA) Marseille, France; 3–5 December 2025.